KSQ-004: Unbiased pair-wise discovery of SOCS1 and Regnase-1 as the top CRISPR/Cas9 dual-edit combination enhancing in vivo TIL potency against solid tumors

Chris Wrocklage, Sharon Lin, Isabelle le Mercier, Caroline Bullock, Louise Cadzow, Anja Hohmann, Solomon Shenker, Katri Sofjan, Leila Williams, Gregory Kryukov, Conor Calnan, Frank Stegmeier, Karrie Wong, Micah J. Benson, and Michael Schlabach

KSQ Therapeutics, Cambridge, MA

Abstract

Background: The application of CRISPR/Cas9-gene-editing to enhance the anti-tumor activity of T cell Adoptive Cell Therapies (ACT) is a promising approach in the treatment of patients with solid tumors. We developed an in vivo CRISPR²™ screening approach and interrogated the top dual-edit combinations enhancing T cell anti-tumor function. We discovered that across all possible dual-edit combinations of T cell targets, inactivation of Regnase-1 and SOCS1 led to the greatest enhancement in anti-tumor T cell potency in vivo. We applied these findings to discover KSQ-004, a Regnase-1/SOCS1 dual-edited human CRISPR/Cas9-engineered TIL (eTIL™) therapy currently under development for therapeutic use.

Methods: We generated randomly paired CRISPR guide libraries (CRISPR²) targeting top hits from previous single-gene Immune CRISPRomics® screens. CRISPR² libraries with over 1200 gene pairs were screened in primary mouse OT1 and PMEL-TCR-Tg-T cells in the relevant syngeneic tumor models. Top dual-edit combinations were then evaluated in the immunotherapy-refractory B16F10 metastatic lung tumor model. The efficacy of the top combo was further evaluated in a mouse TIL model wherein TIL from B16-Ova tumors were expanded and engineered ex vivo and adoptively transferred into tumor bearing hosts for efficacy assessment.

Results: Of the 1200+ combinations tested in the CRISPR² screens, the Regnase-1/SOCS1 combination ranked amongst the top dual-edits, with this combination enhancing T cell infiltration into tumors >3500-fold in comparison to controls. Studies conducted in the checkpoint therapy refractory B16F10 lung metastasis model revealed that Regnase-1/SOCS1 dual-edited PMEL-TCR-Tg-T cells conferred remarkable survival benefit to controls, significantly extending median survival of animals from 21 days to 53 days. Furthermore, Regnase-1+SOCS1-edited mouse TIL isolated and expanded from B16-Ova tumors exerted complete control of tumors upon re-infusion into hosts, suggesting rejuvenation of tumor-experienced TILs by this edit combination. To apply these insights for therapeutic use, we discovered KSQ-004, a human Regnase-1/SOCS1 dual-edited CRISPR/Cas9-engineered TIL (eTIL). Methods were developed to manufacture KSQ-004 from melanoma and NSCLC tumor samples, with eTIL demonstrating robust expansion and viability comparable to unedited control TIL with over 90% knockout of both targets. Importantly, KSQ-004 produced elevated IFNɣ upon autologous tumor stimulation and exerted greater control of tumor spheroids in vitro.

Conclusion: We used a novel CRISPR² screen approach to identify Regnase-1/SOCS1 as the top dual edit combination enhancing T cell function in the tumor microenvironment. We translated these findings to therapeutic use with the discovery of KSQ-004, a dual-edited eTIL therapy that possesses enhanced anti-tumor potency and persistence against solid tumors.

Methods

In Vivo Studies: Generation of mouse KSQ-004 cells (mKSQ-004)

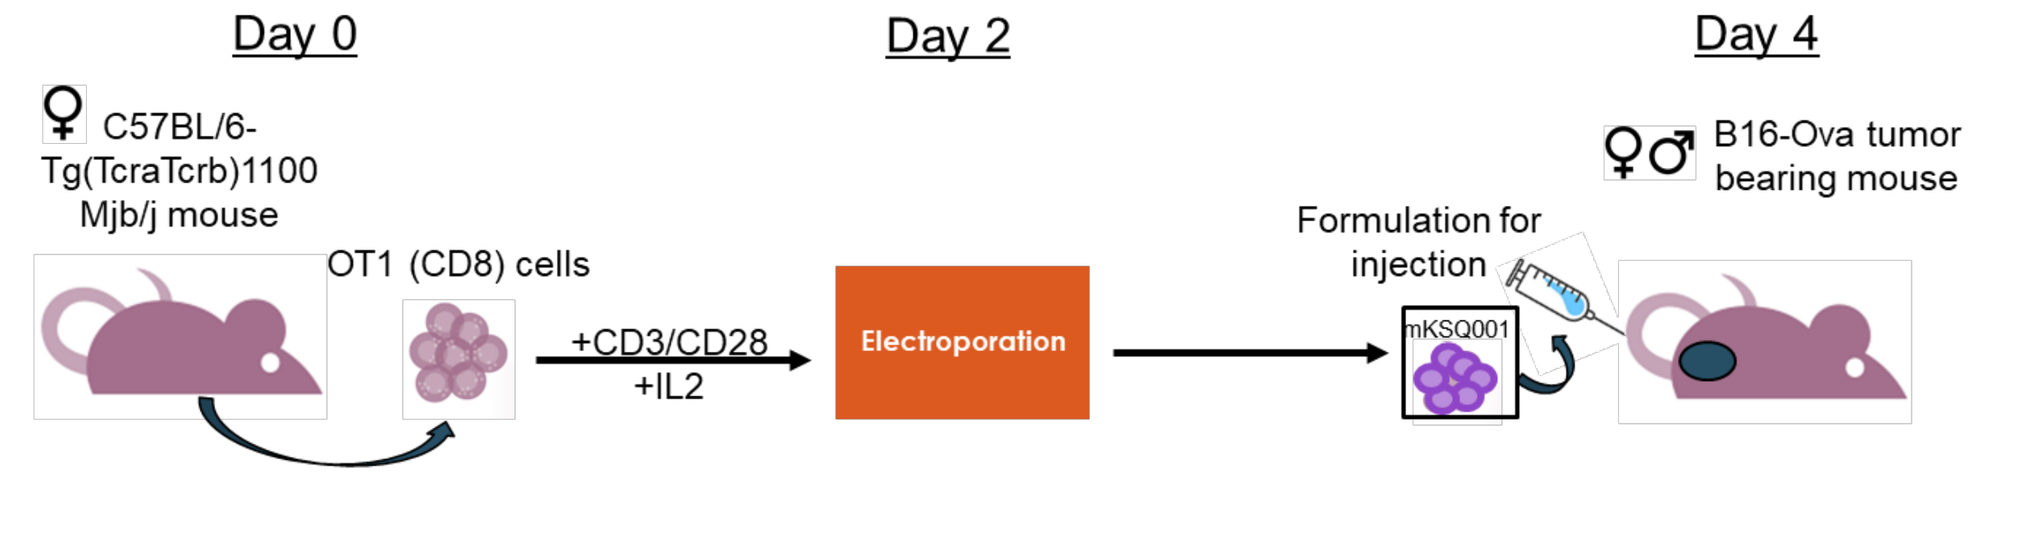
Day 0
Day 2
Day 4
♀ C57BL/6-Tg(TcraTcrb)1100 Mjb/j mouse
OT1 (CD8) cells
+CD3/CD28 +IL2
Electroporation
Formulation for injection
mKSQ001
♀♂ B16-Ova tumor bearing mouse

- To serve as a negative control, T cells are edited with a sgRNA targeting OLF1, an olfactory receptor with no function in T cells

Human KSQ-004:

- Human KSQ-004 from melanoma and NSCLC donors were manufactured. For editing of *SOCS1*, the sgRNA used to manufacture KSQ-001 was used; for editing of *ZC3H12A*, the gene encoding Regnase-1 protein, a top guide was selected based on editing efficiency and minimal off-target editing
- Unedited TILs (No EP), which do not undergo the gene editing step, are generated in each run and serve as controls for in vitro characterization assays
- Both No EP and KSQ-004 were cryopreserved following manufacture; all characterization assays were performed with cryopreserved cells

Figure 1: Discovery and Validation of Regnase-1 and SOCS1 as the Top T cell Dual-Edit Target Combo Enhancing anti-Tumor Function

1a) **CRISPR²: Industrialized Combination T cell Target Discovery**

- 35 top T cell hits selected from prior in vivo genome-wide screening efforts
- 1200 combos evaluated in a single experiment
- Multiple screens conducted in OT1/B16-Ova and PMEL/CRC-gp100 syngeneic models

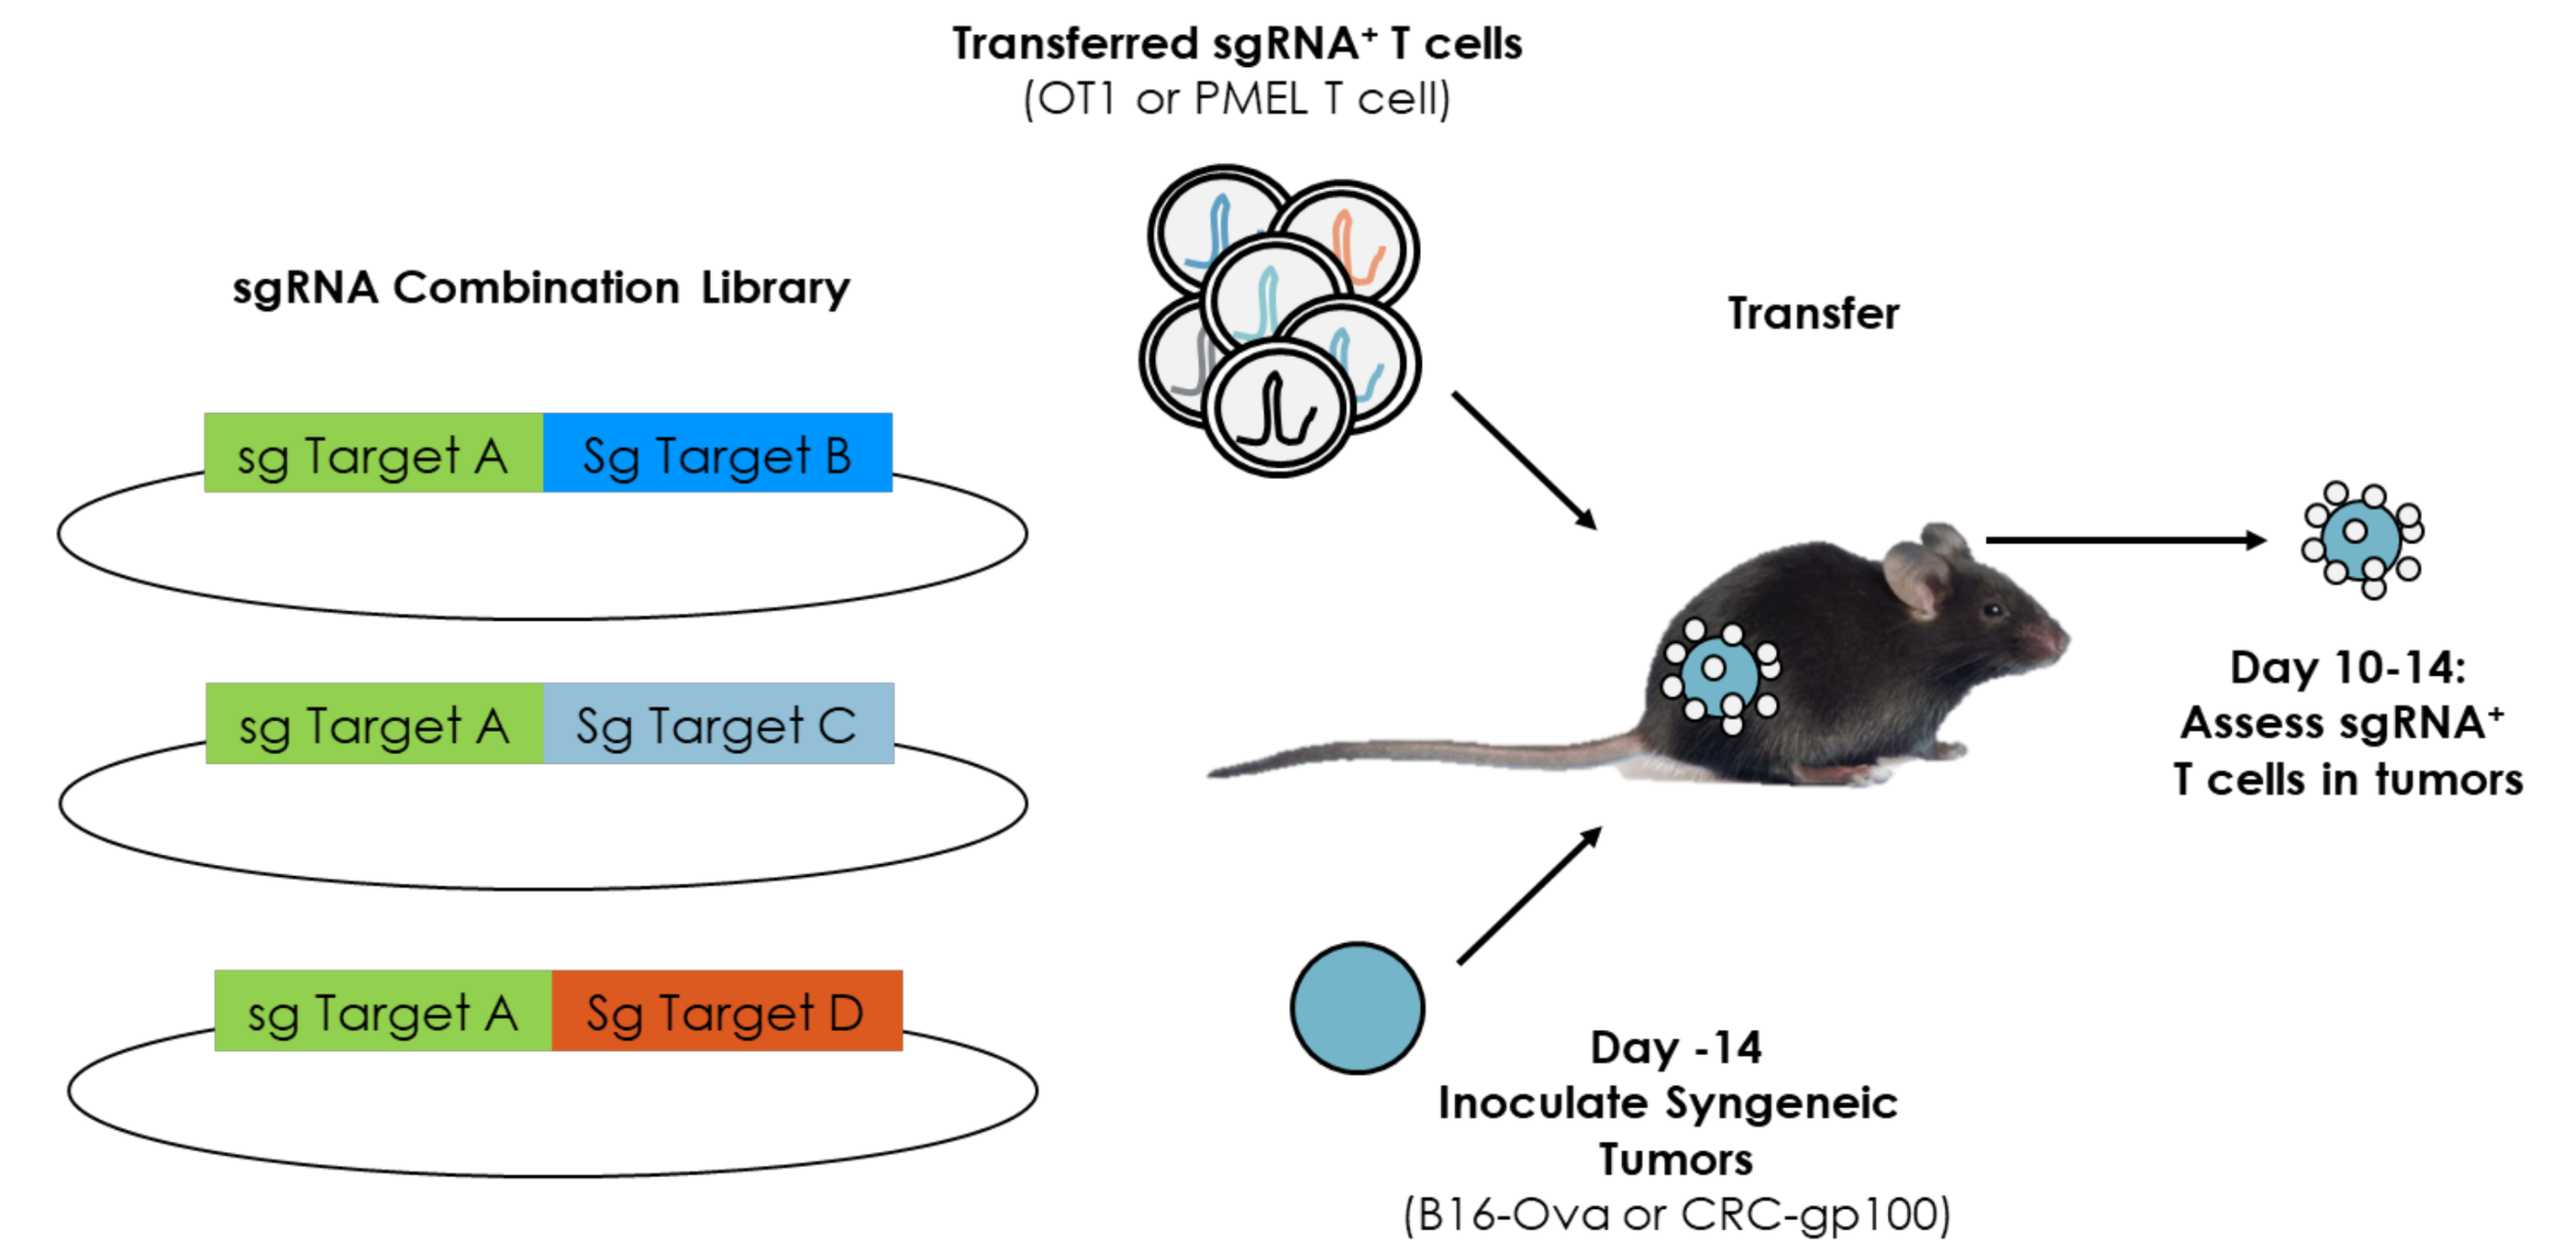
Transferred sgRNA⁺ T cells
(OT1 or PMEL T cell)
sgRNA Combination Library
sg Target A
Sg Target B
sg Target A
Sg Target C
sg Target A
Sg Target D
Transfer
Day 10-14:
Assess sgRNA⁺
T cells in tumors
Day -14
Inoculate Syngeneic
Tumors
(B16-Ova or CRC-gp100)

1b) **CRISPR²: In vivo T cell combination screen identifies Regnase-1 and SOCS1 as the top dual-edit combo boosting T cell accumulation into solid tumors**

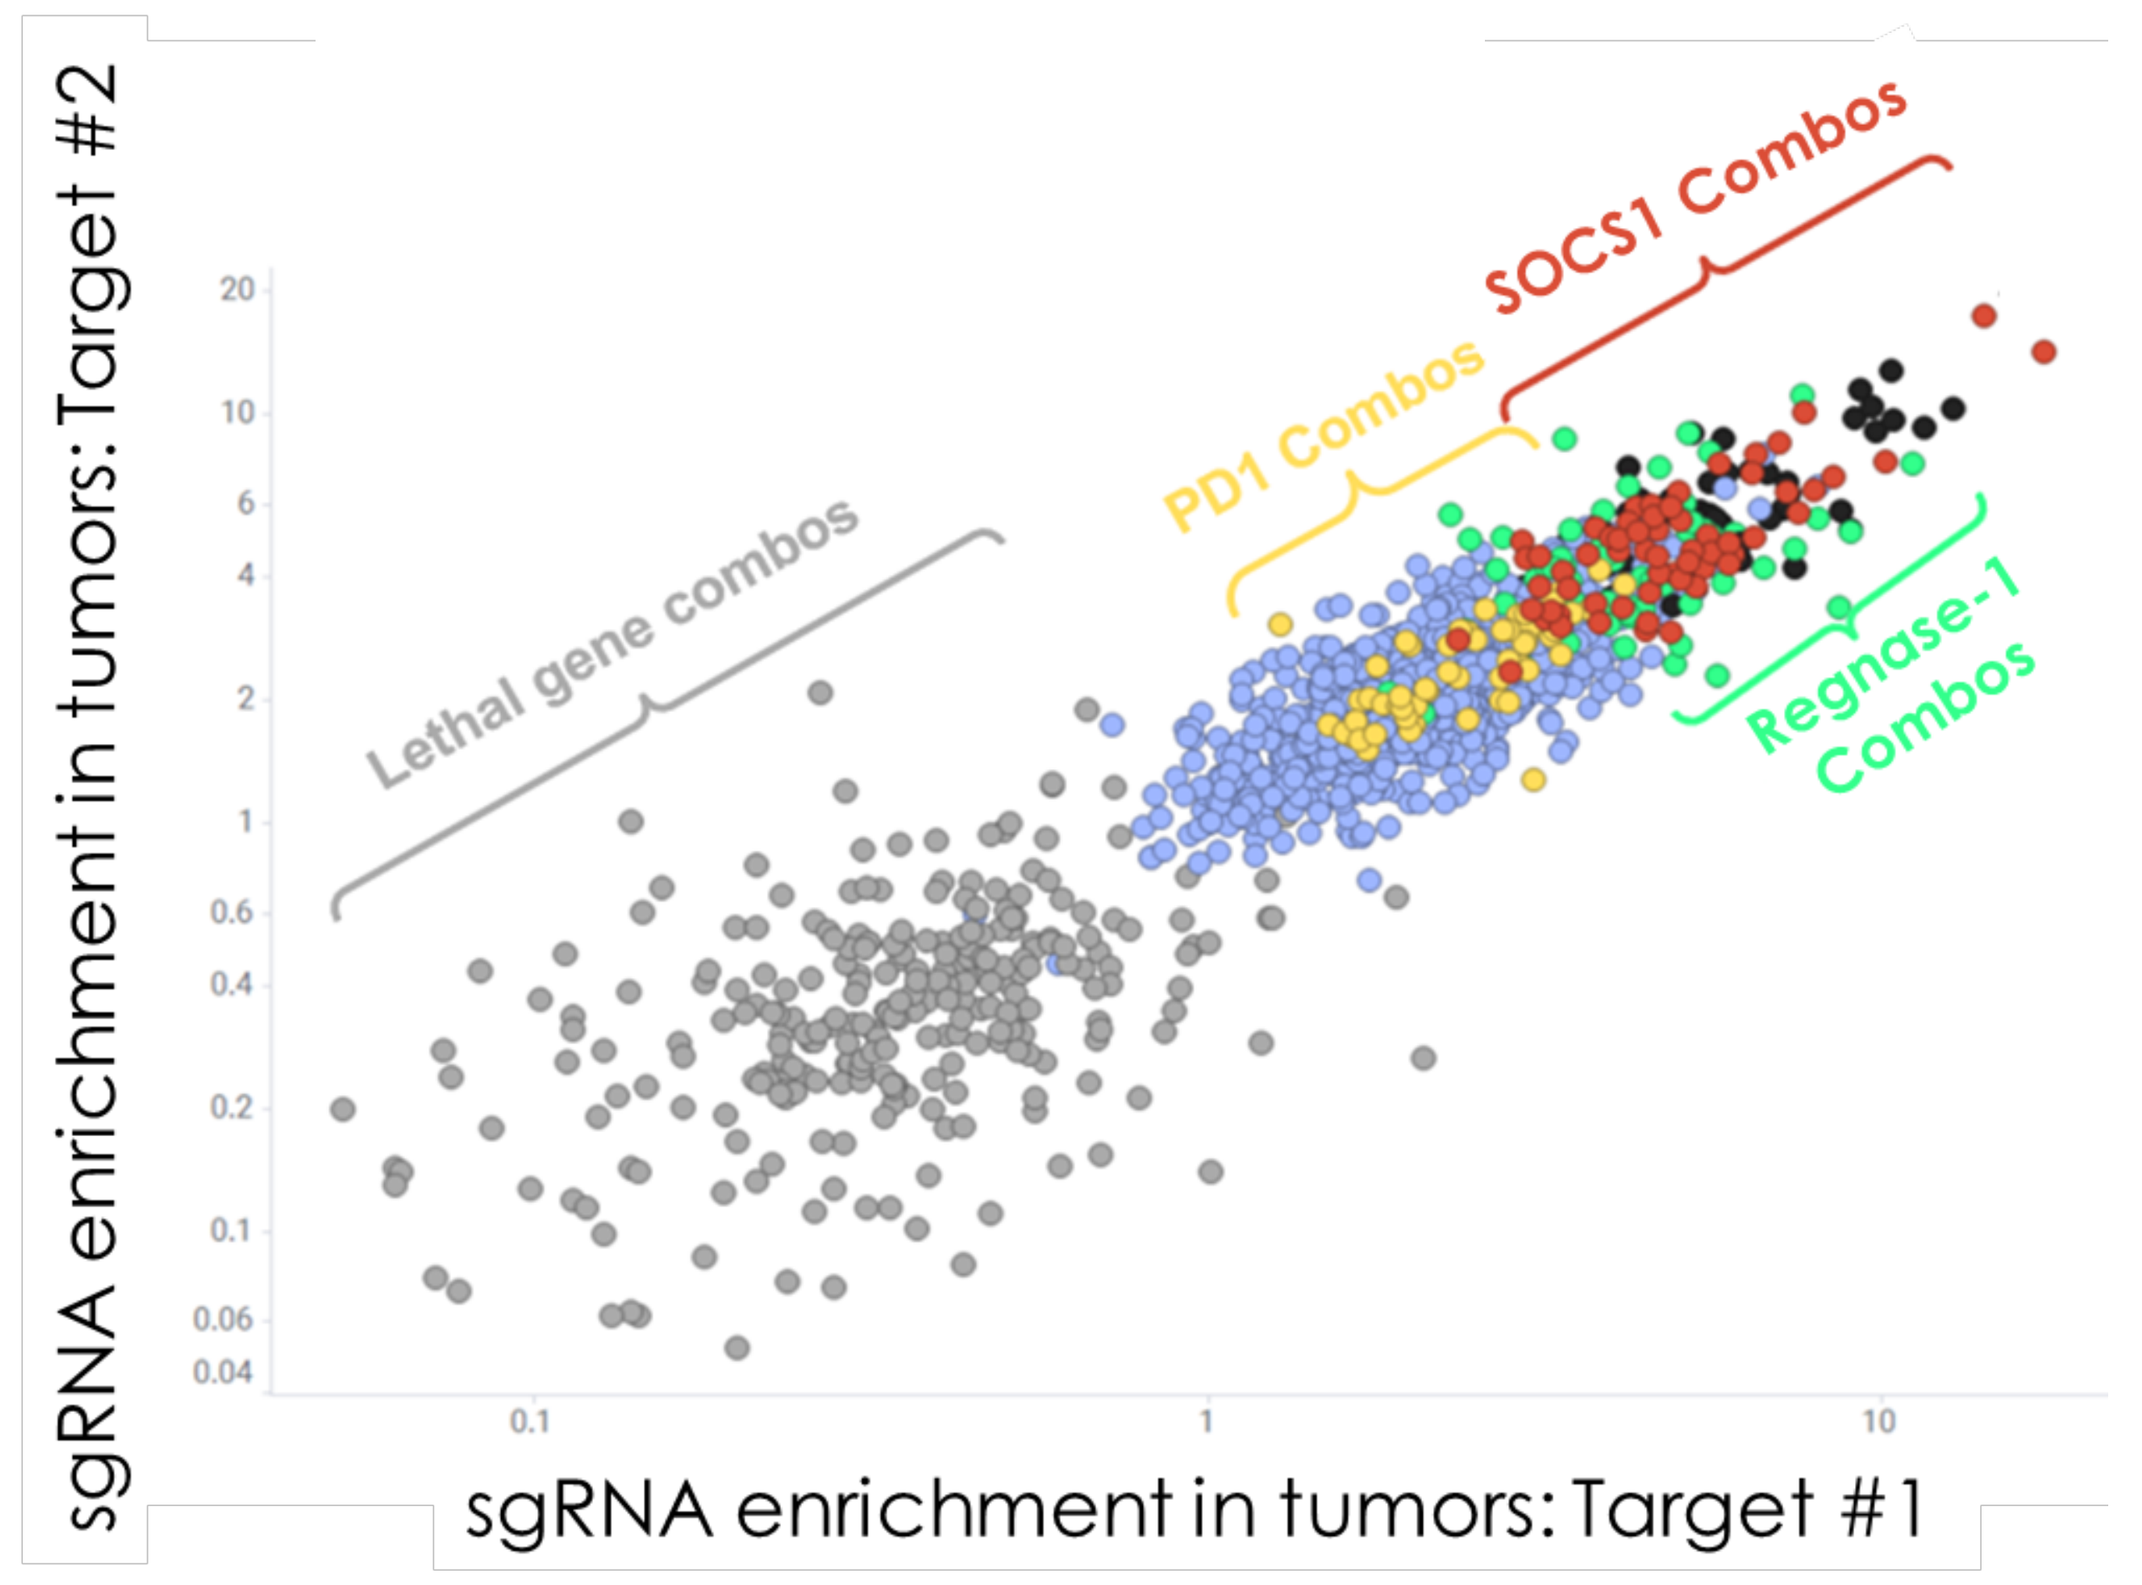
sgRNA enrichment in tumors: Target #2
sgRNA enrichment in tumors: Target #1
Lethal gene combos
PD1 Combos
SOCS1 Combos
Regnase-1 Combos

- In vivo T cell combo screen: measures abundance of T cells in tumor
- Screen performed in both Mouse OT1/B16-Ova and PMEL / CRC-gp100 syngeneic models
- Dual inactivation of *Zc3h12a*, which encodes the Regnase-1 protein, and *Socs1* genes increased OT1 T cell infiltration in tumors >3500-fold (vs 50-fold for PD1)
- In general, inactivation of PD-1 showed limited combo activity

1c) **Validation of Regnase-1 and SOCS1 as the T cell top dual-edit combo in a treatment-refractory PMEL / B16F10 Lung Metastasis Model**

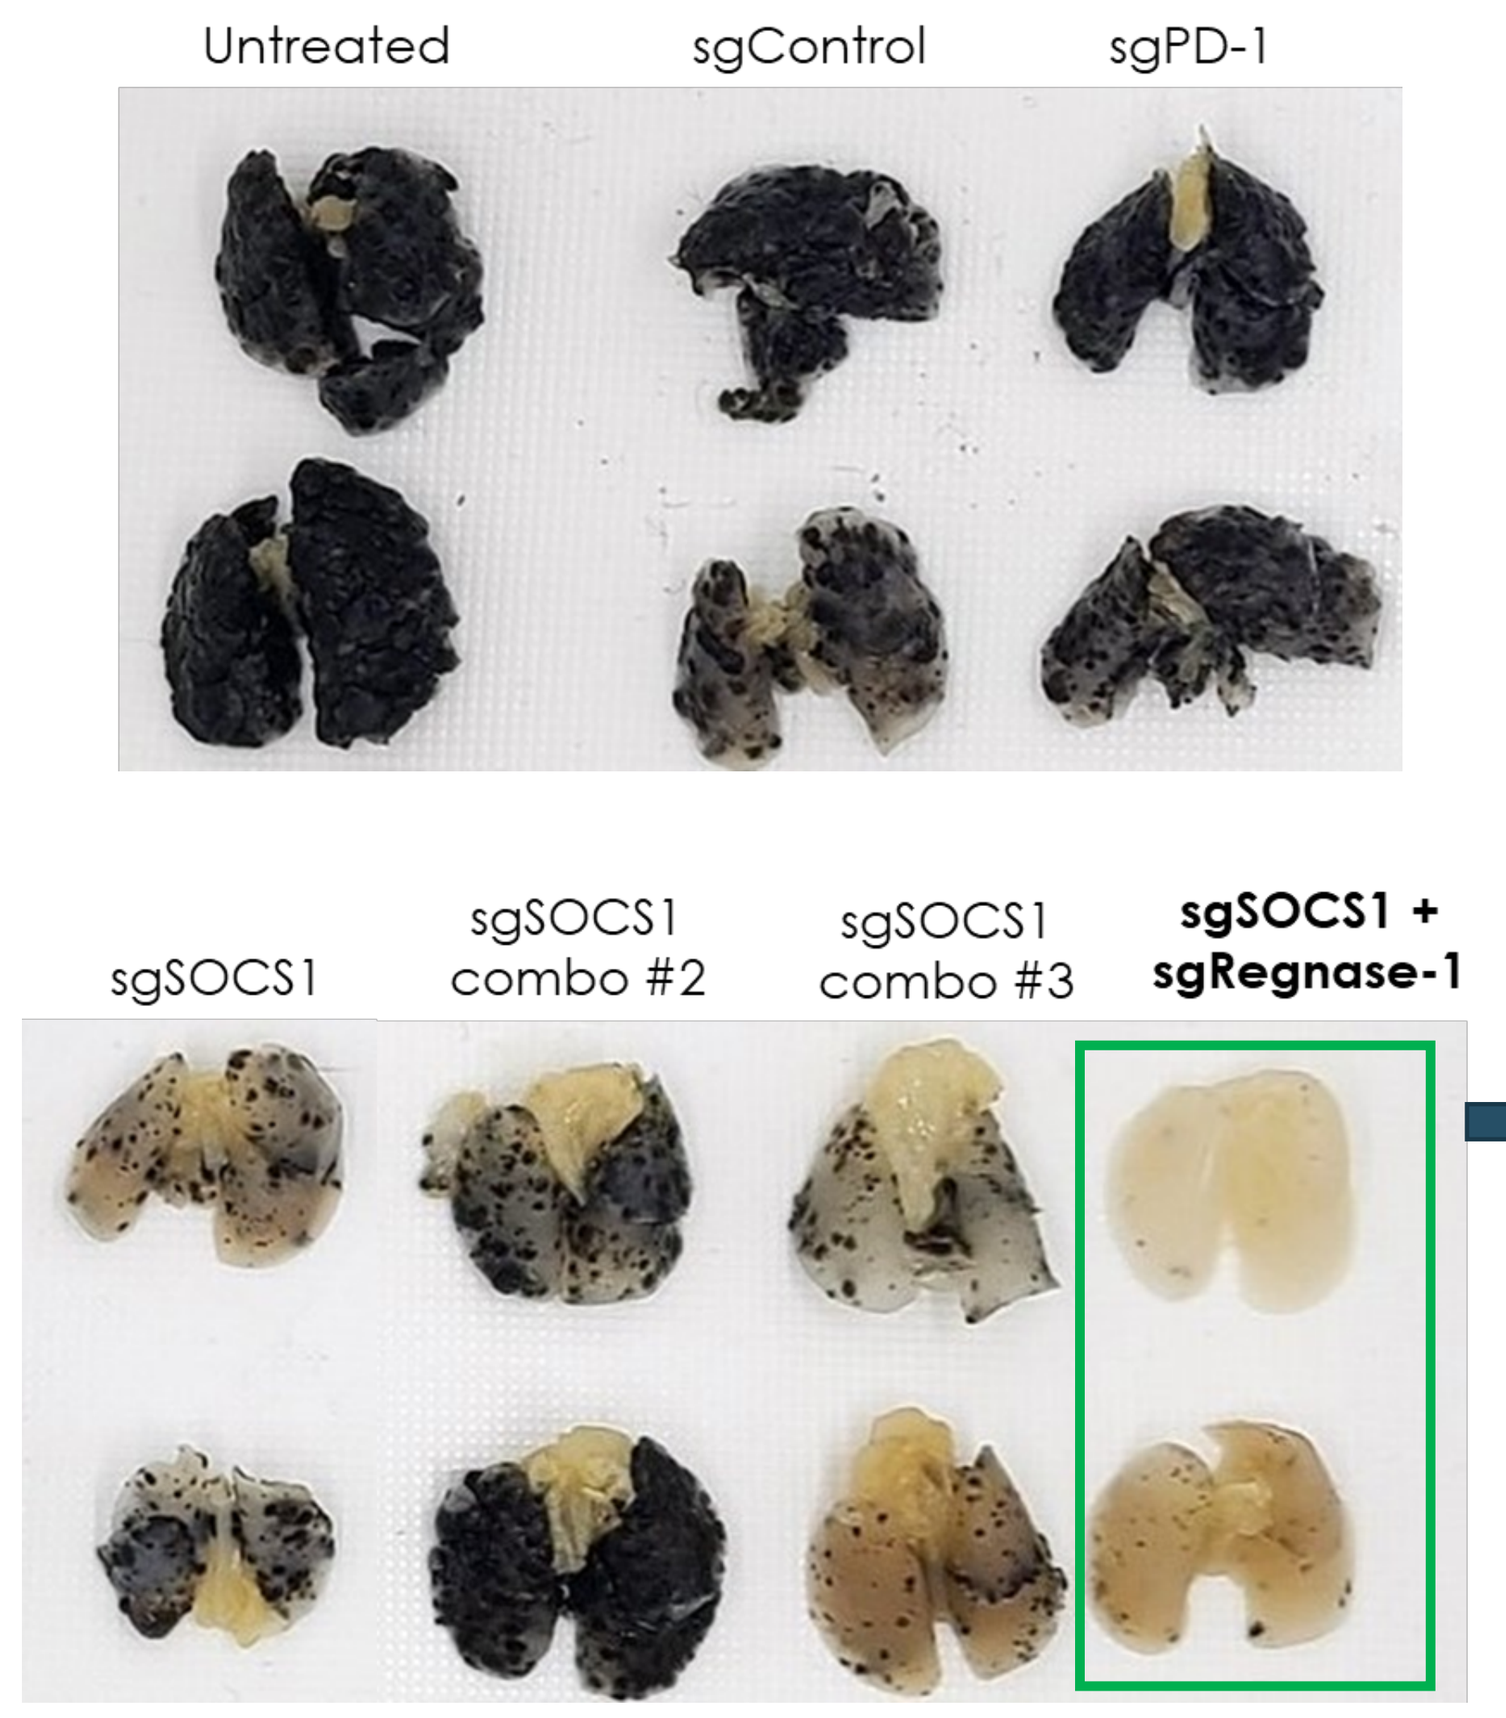
Untreated
sgControl
sgPD-1
sgSOCS1
sgSOCS1 combo #2
sgSOCS1 combo #3
sgSOCS1 + sgRegnase-1
Survival

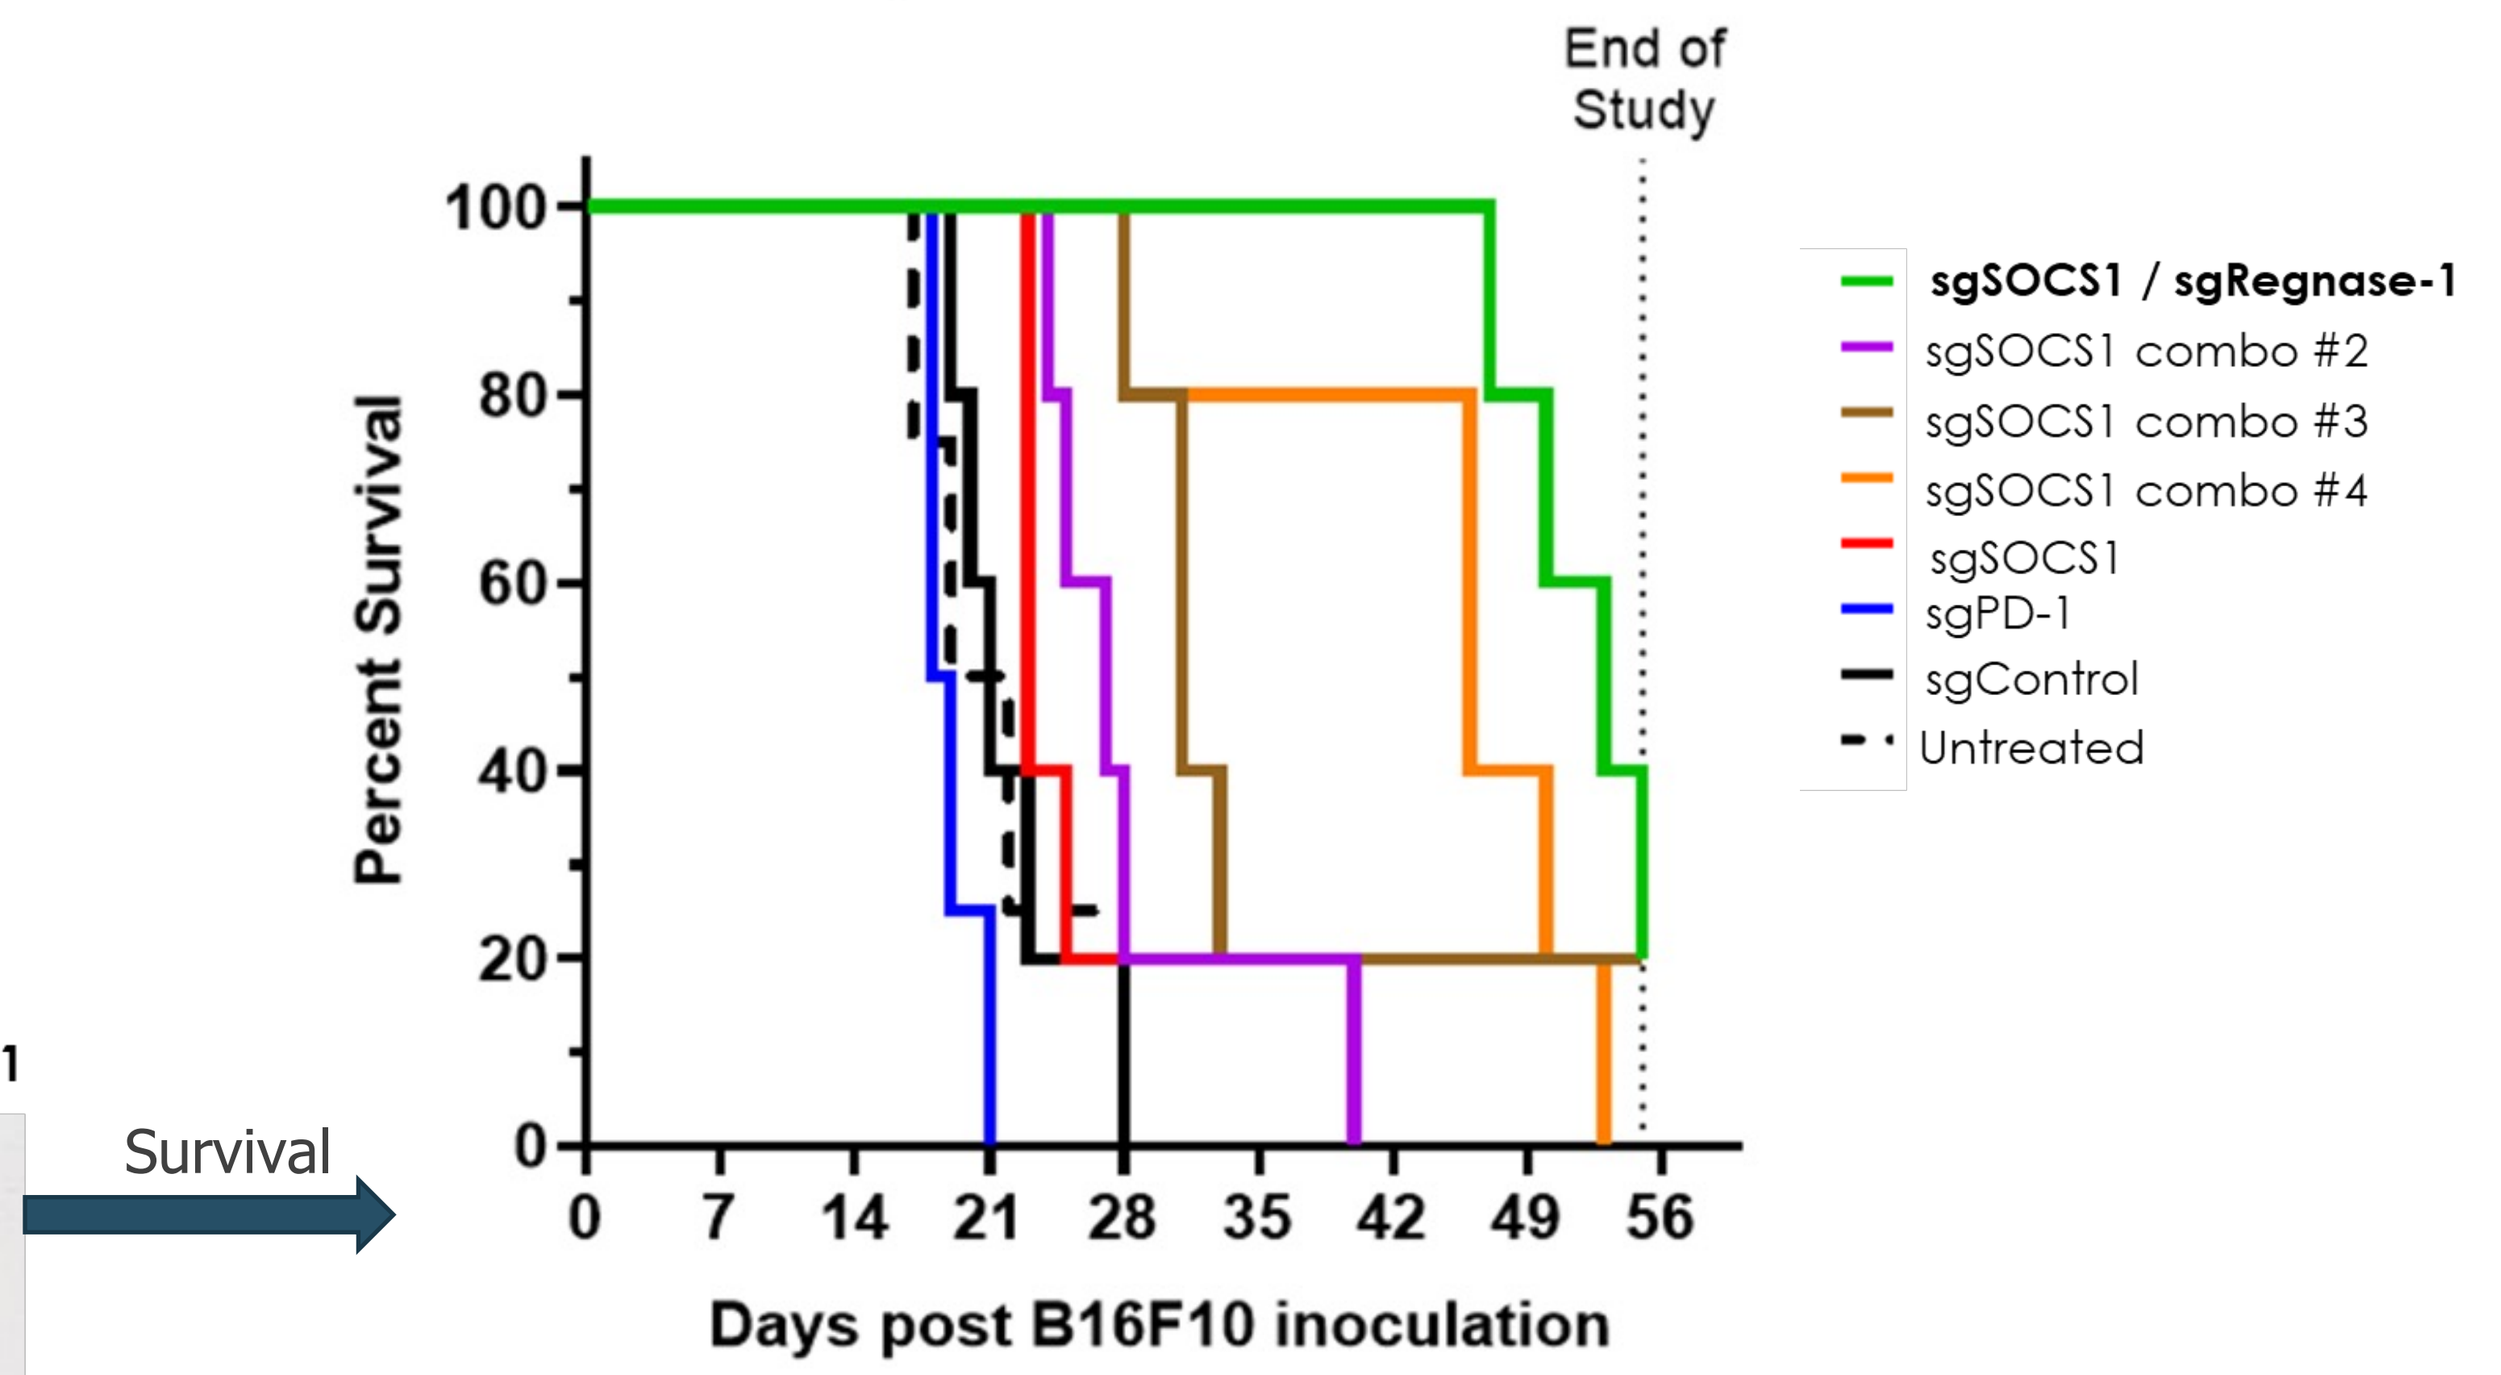
End of Study
Percent Survival
Days post B16F10 inoculation
sgSOCS1 / sgRegnase-1
sgSOCS1 combo #2
sgSOCS1 combo #3
sgSOCS1 combo #4
sgSOCS1
sgPD-1
sgControl
Untreated

- 1x10⁶ CRISPR/Cas9-engineered PMEL CD8 T cells were adoptively transferred into mice at 3 days following IV injection of B16F10 tumor cells
- Out of all target combinations tested, a Socs1 / Regnase-1 dual-edit combo led to the best overall survival, with this combo selected for KSQ-004, and dual-edited eTIL product

Figure 2: Robust anti-Tumor Activity of murine KSQ-004 in a mouse TIL Model

2a) **Mouse TIL Model**

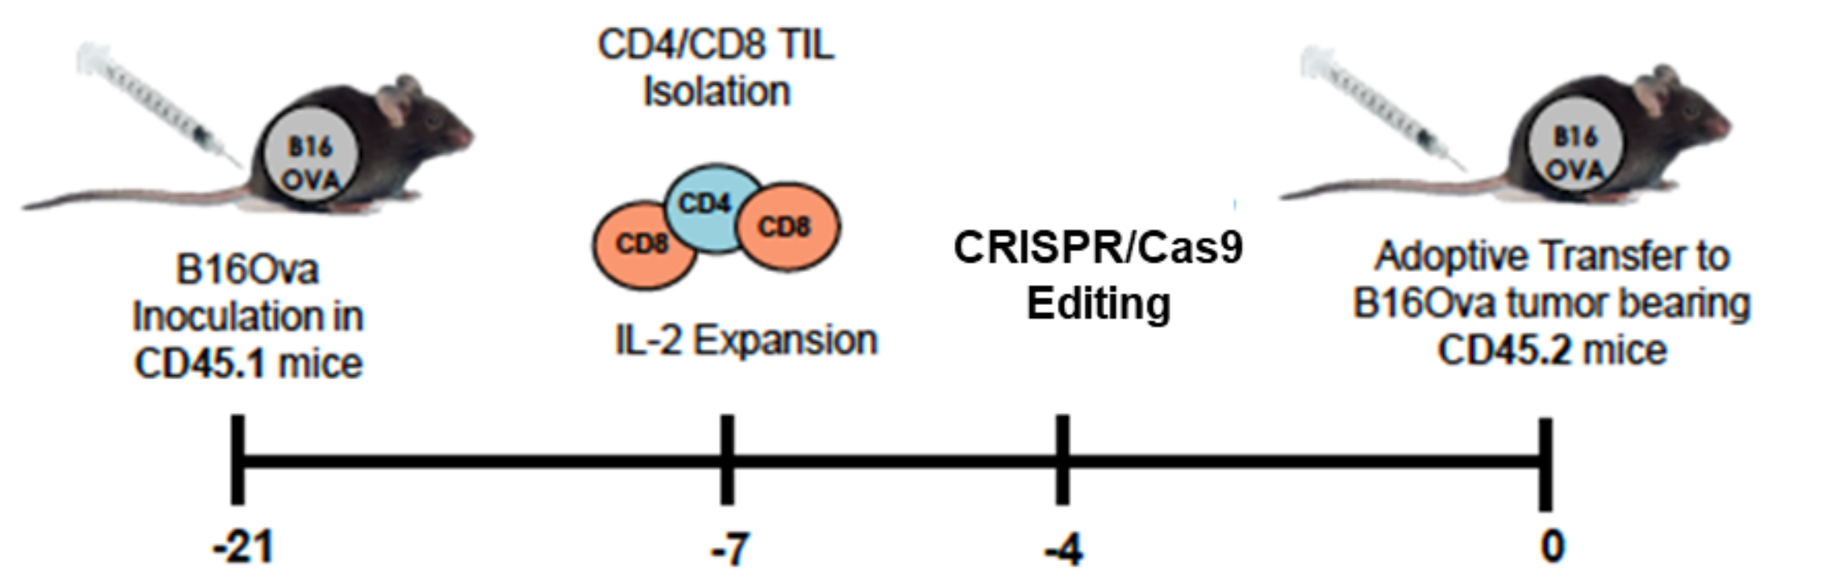
B16Ova Inoculation in CD45.1 mice
CD4/CD8 TIL Isolation
IL-2 Expansion
CRISPR/Cas9 Editing
Adoptive Transfer to B16Ova tumor bearing CD45.2 mice
-21
-7
-4
0

- Murine TIL were isolated from B16-Ova tumors, expanded and engineered to knockout Regnase-1 and Socs1 ex-vivo using CRISPR/Cas9 (murine KSQ-004)
- Murine TIL were then adoptively transferred into B16-Ova tumor bearing animals

2b) **Murine KSQ-004 drive complete tumor regressions**

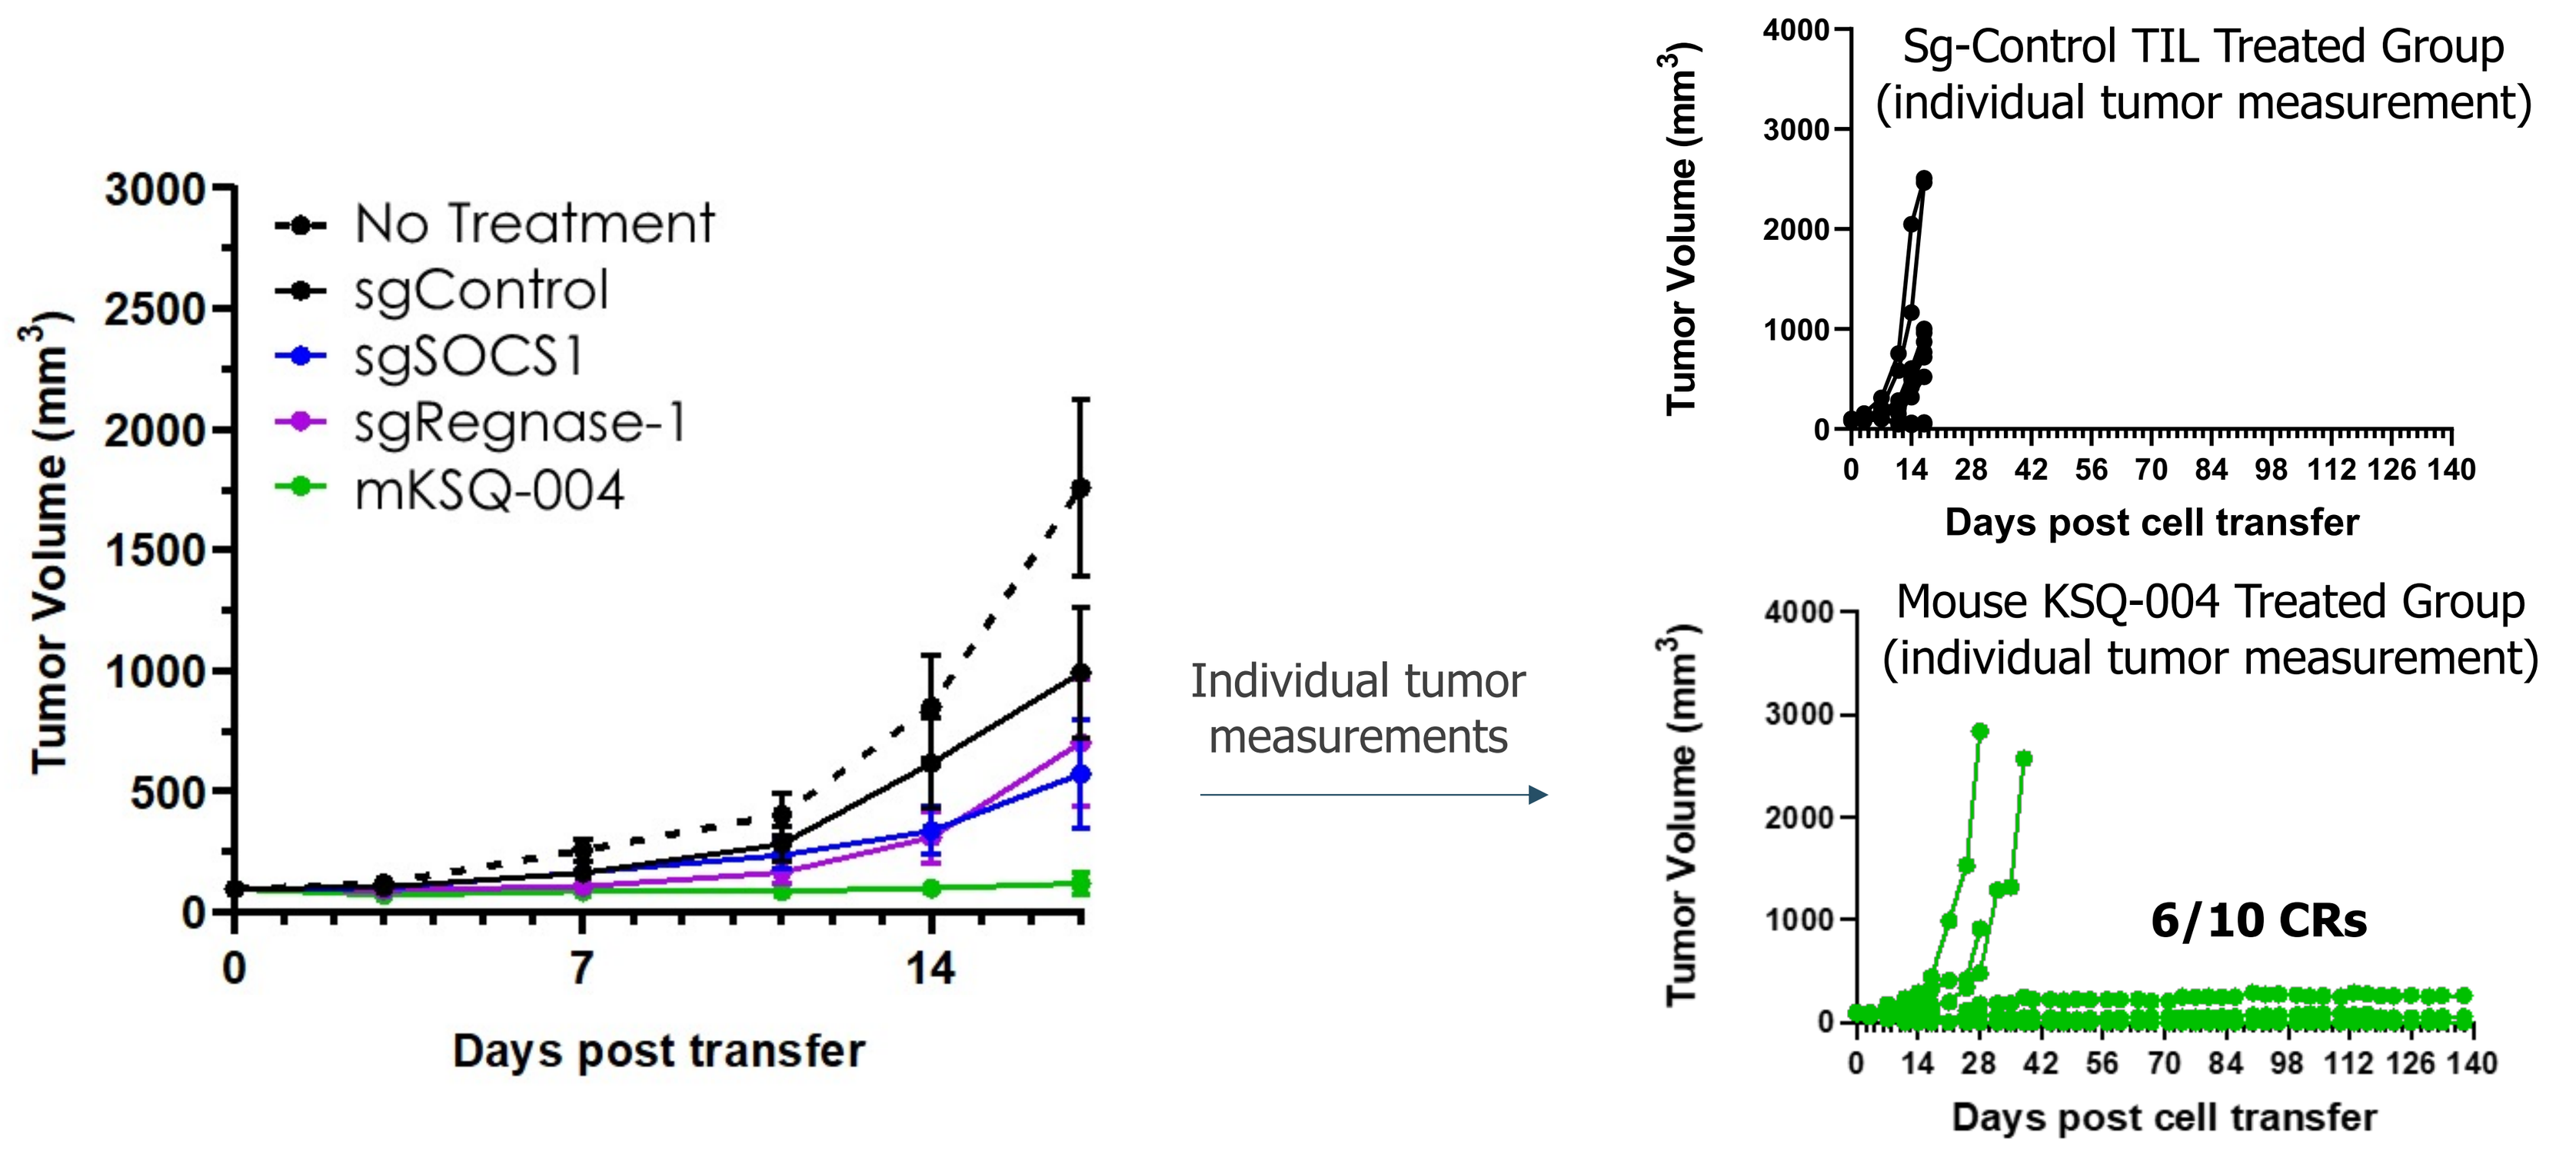
Tumor Volume (mm³)
Days post transfer
No Treatment
sgControl
sgSOCS1
sgRegnase-1
mKSQ-004
Individual tumor measurements
Sg-Control TIL Treated Group
(individual tumor measurement)
Mouse KSQ-004 Treated Group
(individual tumor measurement)
6/10 CRs
Days post cell transfer

Figure 3: Characterization of Human KSQ-004, a Regnase-1 / SOCS1 Dual Edited eTIL™ Product

KSQ-004: An autologous, dual-gene edited tumor-infiltrating lymphocyte (TIL) cell product wherein Cas9/sgRNAs targeting the Regnase-1 and SOCS1 genes are electroporated (EP) into TIL

3a) **Manufacture of human KSQ-004: comparable expansion and viability with unedited TIL**

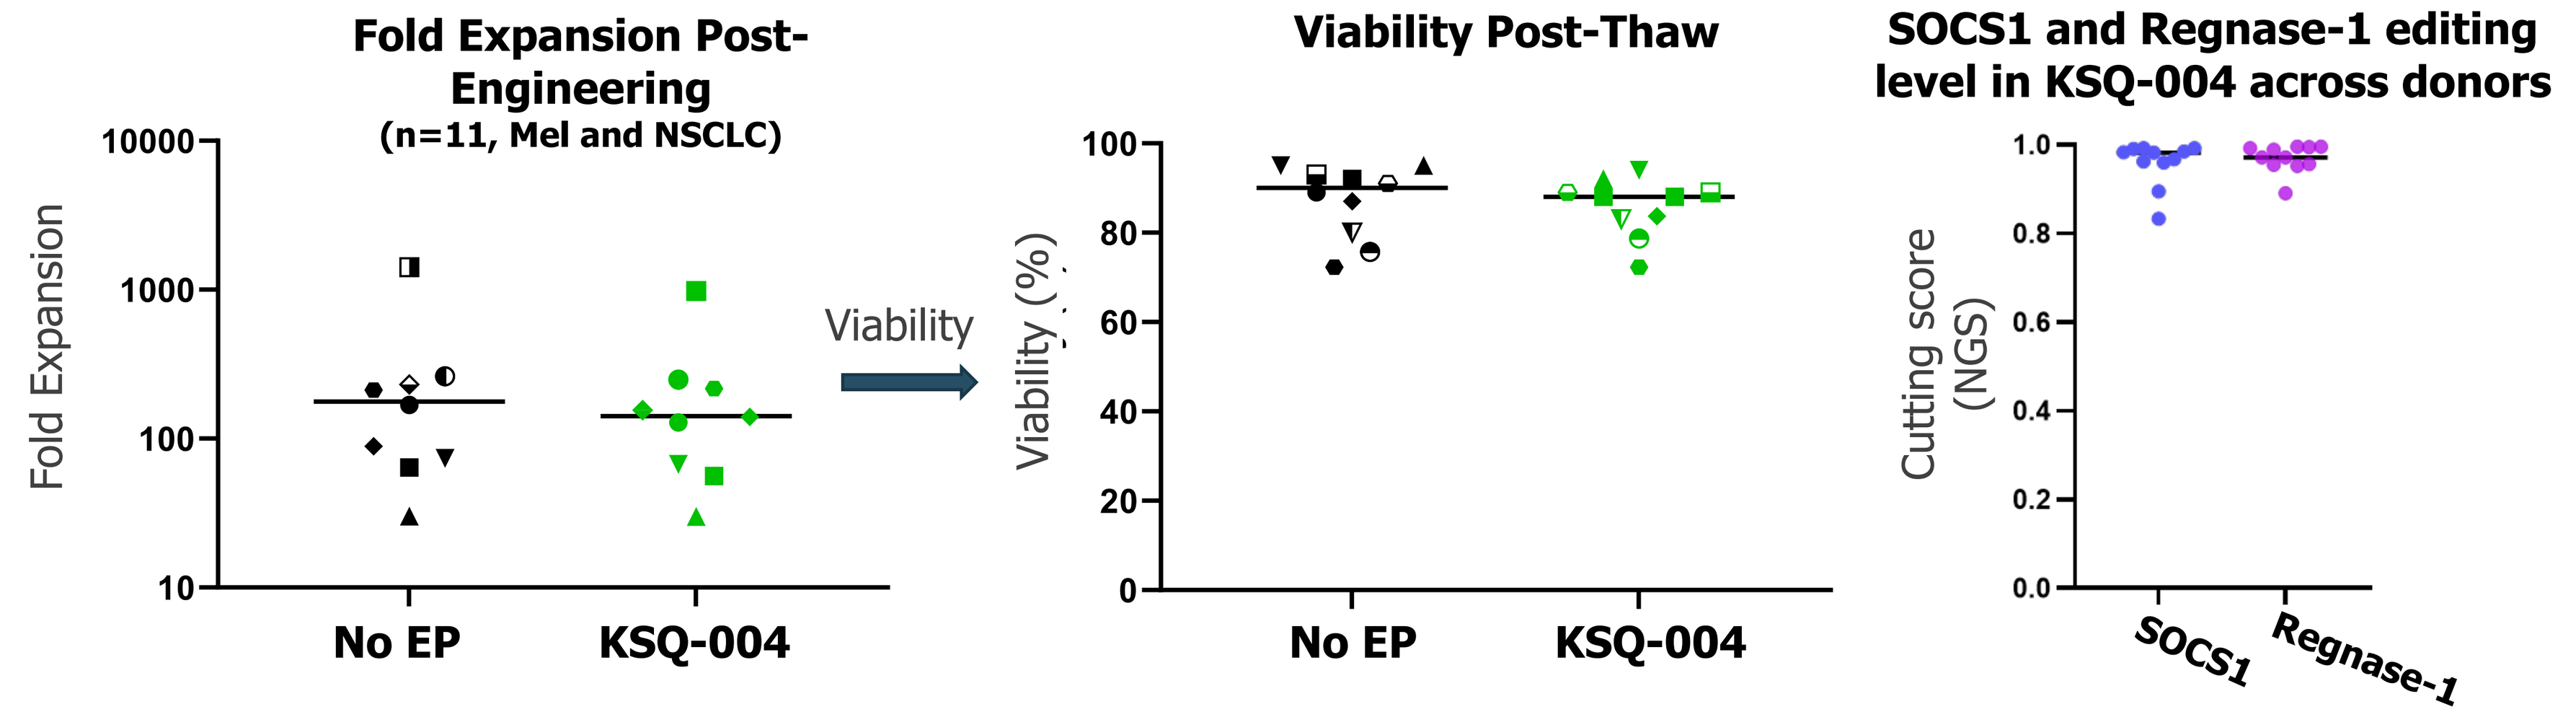
Fold Expansion Post-Engineering
(n=11, Mel and NSCLC)
Fold Expansion
No EP
KSQ-004
Viability
Viability Post-Thaw
Viability (%)
SOCS1 and Regnase-1 editing level in KSQ-004 across donors
Cutting score (NGS)
SOCS1
Regnase-1

3b) **Engineering and characterization of KSQ-004: Enhanced *in vitro* anti-tumor activity**

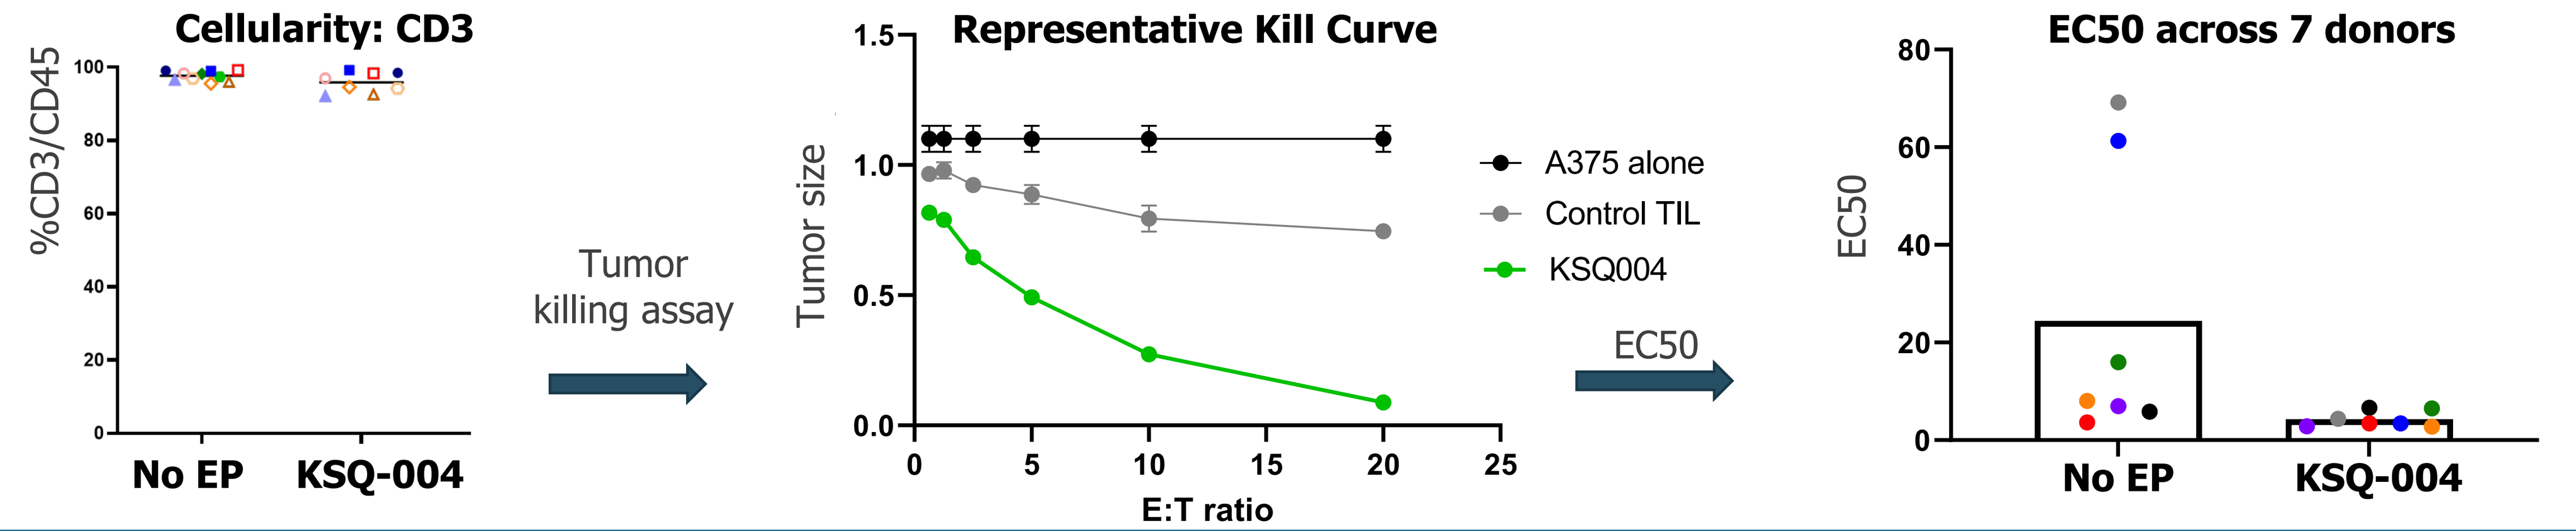
Cellularity: CD3
%CD3/CD45
No EP
KSQ-004
Tumor killing assay
Representative Kill Curve
Tumor size
E:T ratio
A375 alone
Control TIL
KSQ004
EC50
EC50 across 7 donors

Conclusions

- SOCS1 and Regnase-1 were discovered and validated as the top dual edit combo in enhancing anti-tumor T cell function
- KSQ-004, a SOCS1 and Regnase-1 dual edited TIL product, can be manufactured and demonstrates enhanced anti-tumor function in vitro