Comprehensive Identification of Novel Therapeutic Targets for Treatment of PD-1 Resistant Solid Tumors via a Genome-Scale CRISPR/Cas9 *in vivo* T-cell Screen

Isabelle Le Mercier, Jason J. Merkin, Sean Keegan, Conor Calnan, Anja F. Hohmann, Nick Colletti, Eric Fagerberg, Sol Shenker, Caroline Bullock, Chris Wrocklage, Noah Tubo, Tianlei Xu, Matt Noyes, Rami Rahal, Sean Arlauckas, Aria Pearlman Morales, Frank Stegmeier, Louise Cadzow, Mike Schlabach, Gregory V. Kryukov, Micah J. Benson

KSQ Therapeutics, Cambridge, MA

Abstract

Although immunotherapy with PD-1/PD-L1 antagonists has significantly advanced patient care, the majority of cancer patients currently do not benefit from checkpoint inhibitor therapies.

To identify novel targets for the treatment of PD-1 insensitive cancers, we developed a novel Immune-CRISPRomics® platform that enabled genome-wide CRISPR/Cas9 screens in primary T cells in an in vivo setting. Using both OT1 and PMEL TCR-Tg CD8 T cells, we screened for novel targets in both PD-1 sensitive and insensitive syngeneic models. Notably, these screens identified clinically active molecules, such as PD-1 and also predicted recent clinical failures. In addition, we identified multiple targets that enhanced anti-tumor T-cell function similar to or better than PD-1 as a monotherapy, with top targets scoring across models.

The anti-tumor activity of targets was assessed across a collection of PD-1 sensitive and PD-1 refractory syngeneic tumor models. IO-7, CT-1 and IO-12 were found to possess robust activity across multiple PD-1 refractory models. Mechanistic follow-up studies demonstrate that IO-7 inhibition leads to an expansion of central memory T-cell subsets, which have been implicated in driving the durable clinical response of checkpoint inhibitors.

We describe a genome-wide in vivo T-cell CRISPR/Cas9 screen and identify multiple therapeutic targets that present promising therapeutic opportunities for the treatment of checkpoint refractory solid tumors.

Methods

In vivo T-cell screen : CD8 T cell were isolated from OT1 or PMEL x Cas9 Transgenic mice, transduced with a sgRNA library and adoptively transferred into B16-OVA or gp100 CRC tumor bearing mice. After in vivo expansion, tumor were collected, gDNA was isolated and sgRNA accumulation was evaluated by NGS.

In vivo Target validation : B16-OVA melanoma or gp100 CRC cancer cells were injected s.c. into C57BL/6J mice. Mice were randomized for tumor volume (approx. mean 100mm³ or 450mm³) and either control, PD-1, IO-7, IO-12 or CT-1 edited OT1 or PMEL T cells were injected i.v. Tumor volume was measured twice a week. In Figure 4, B16-F10 cells were injected i.v. into C57BL/6J mice. Three days later, control, PD-1 or IO-7 edited PMEL T cells were injected i.v. Twelve days following adoptive transfer of T cells, lungs were collected and images were taken.

In vivo MOA : B16-OVA melanoma cells were injected s.c. into CD45.1 mice. Mice were randomized for tumor size, and either control, PD-1 or IO-7 edited OT1 T cells were injected i.v. Five days post adoptive-transfer of T cells, tumors were harvested, TILs were isolated and analyzed by Single-Cell RNA-seq (10X Genomics).

Figure 1: Identification of Novel Targets by CRISPR/Cas9 *in vivo* T- cell screen

Experimental schematic of OT1 / B16-OVA in vivo T cell CRISPR screen

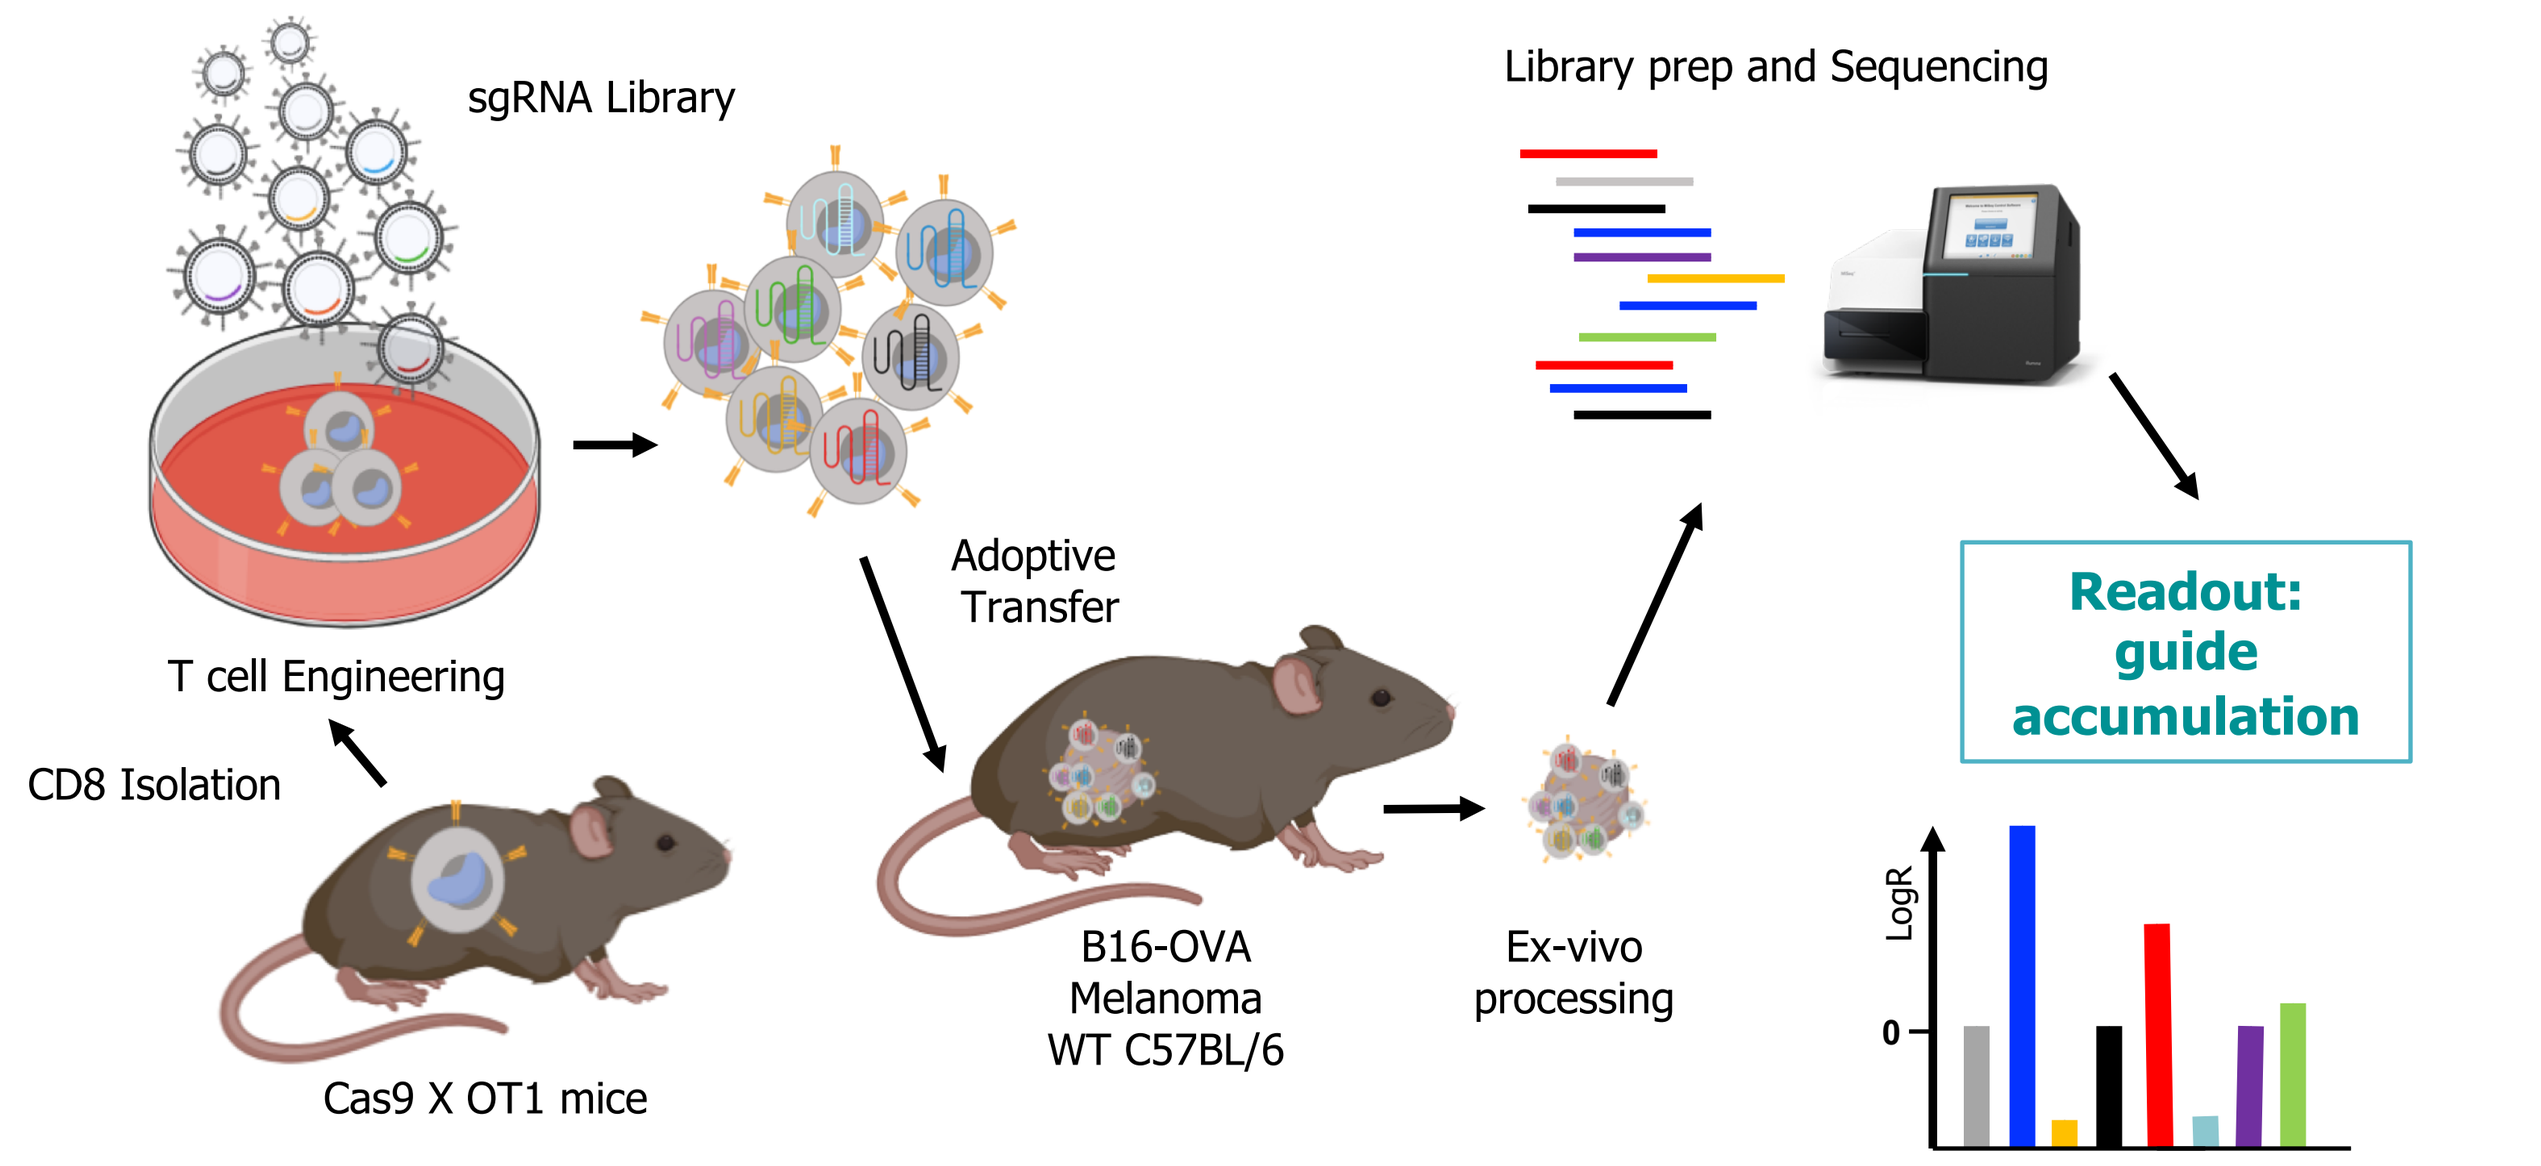
sgRNA Library
T cell Engineering
CD8 Isolation
Cas9 X OT1 mice
Adoptive Transfer
B16-OVA Melanoma WT C57BL/6
Ex-vivo processing
Library prep and Sequencing
Readout: guide accumulation
LogR
0

Screen is robustly powered and validated by multiple quality-control checks

Essential Gene Depletion

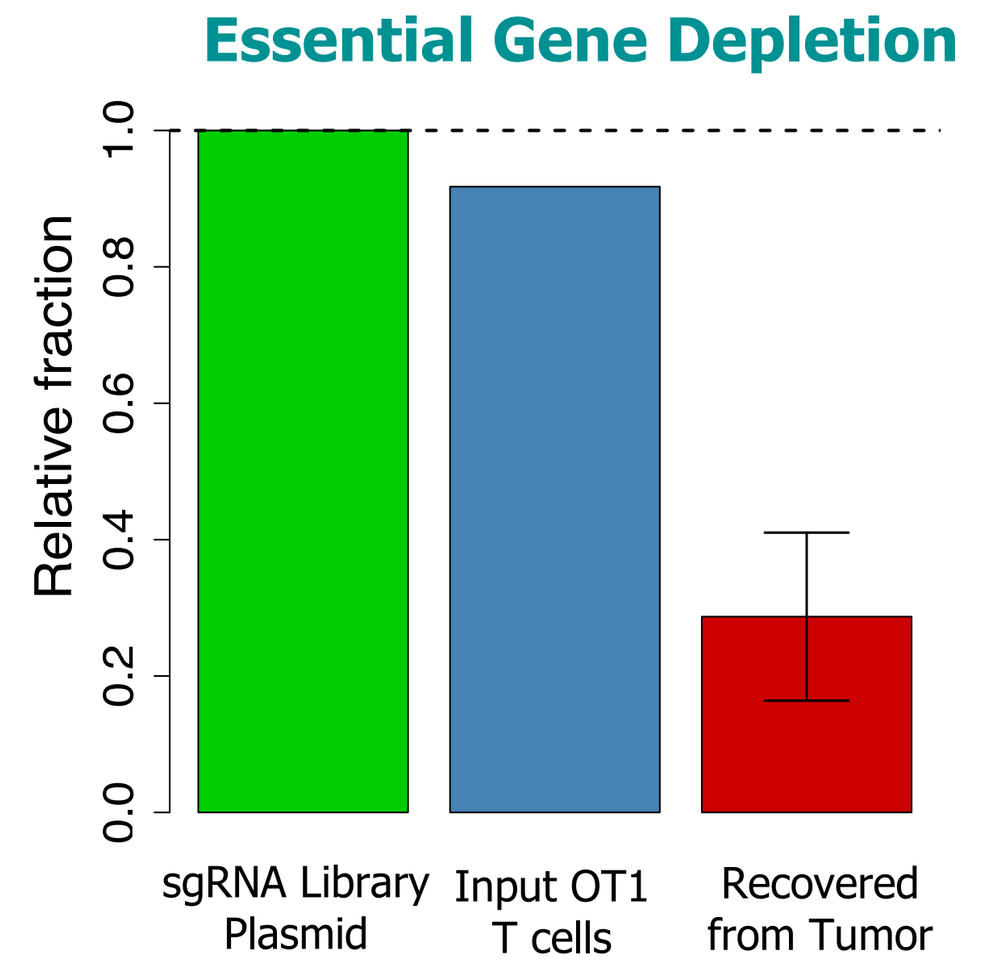
Relative fraction
0.0
0.2
0.4
0.6
0.8
1.0
sgRNA Library Plasmid
Input OT1 T cells
Recovered from Tumor

sgRNA Recovery

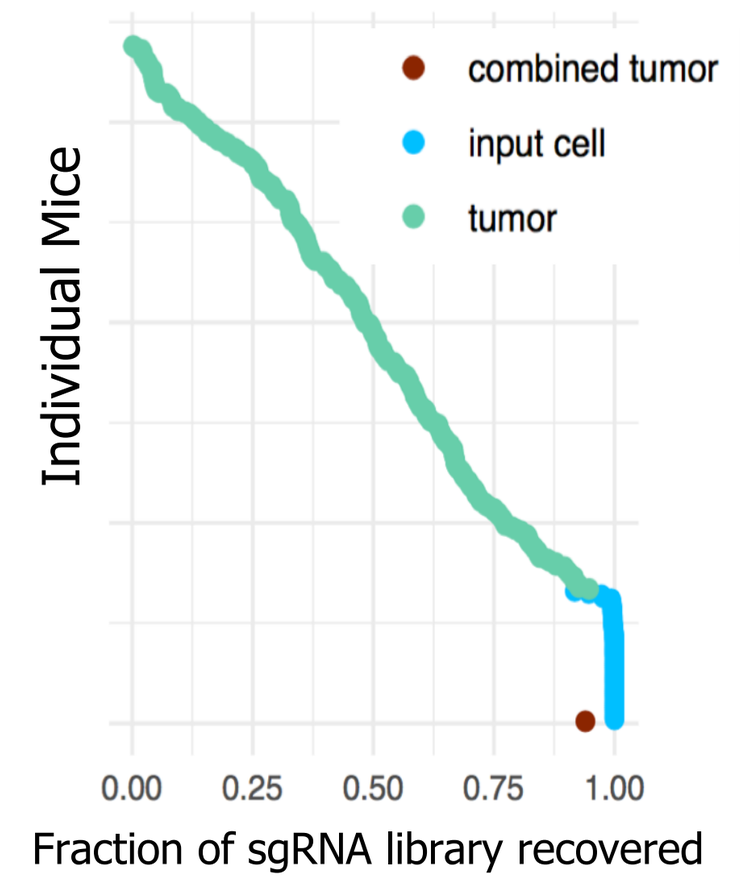
combined tumor
input cell
tumor
Individual Mice
0.00
0.25
0.50
0.75
1.00
Fraction of sgRNA library recovered

Expression by T cells

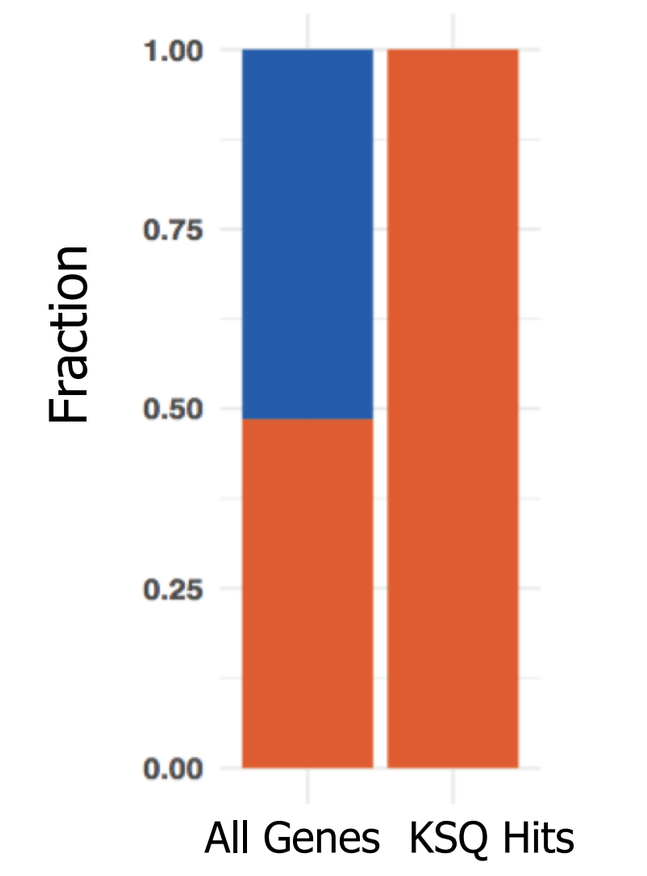
Fraction
0.00
0.25
0.50
0.75
1.00
All Genes
KSQ Hits

- Robust depletion of essential genes in tumor infiltrating T cells
- Robust sgRNA recovery from individual tumors
- All KSQ hits are expressed at the mRNA level by T cells

Genome-Scale in vivo Screen Identifies Novel T cell Targets

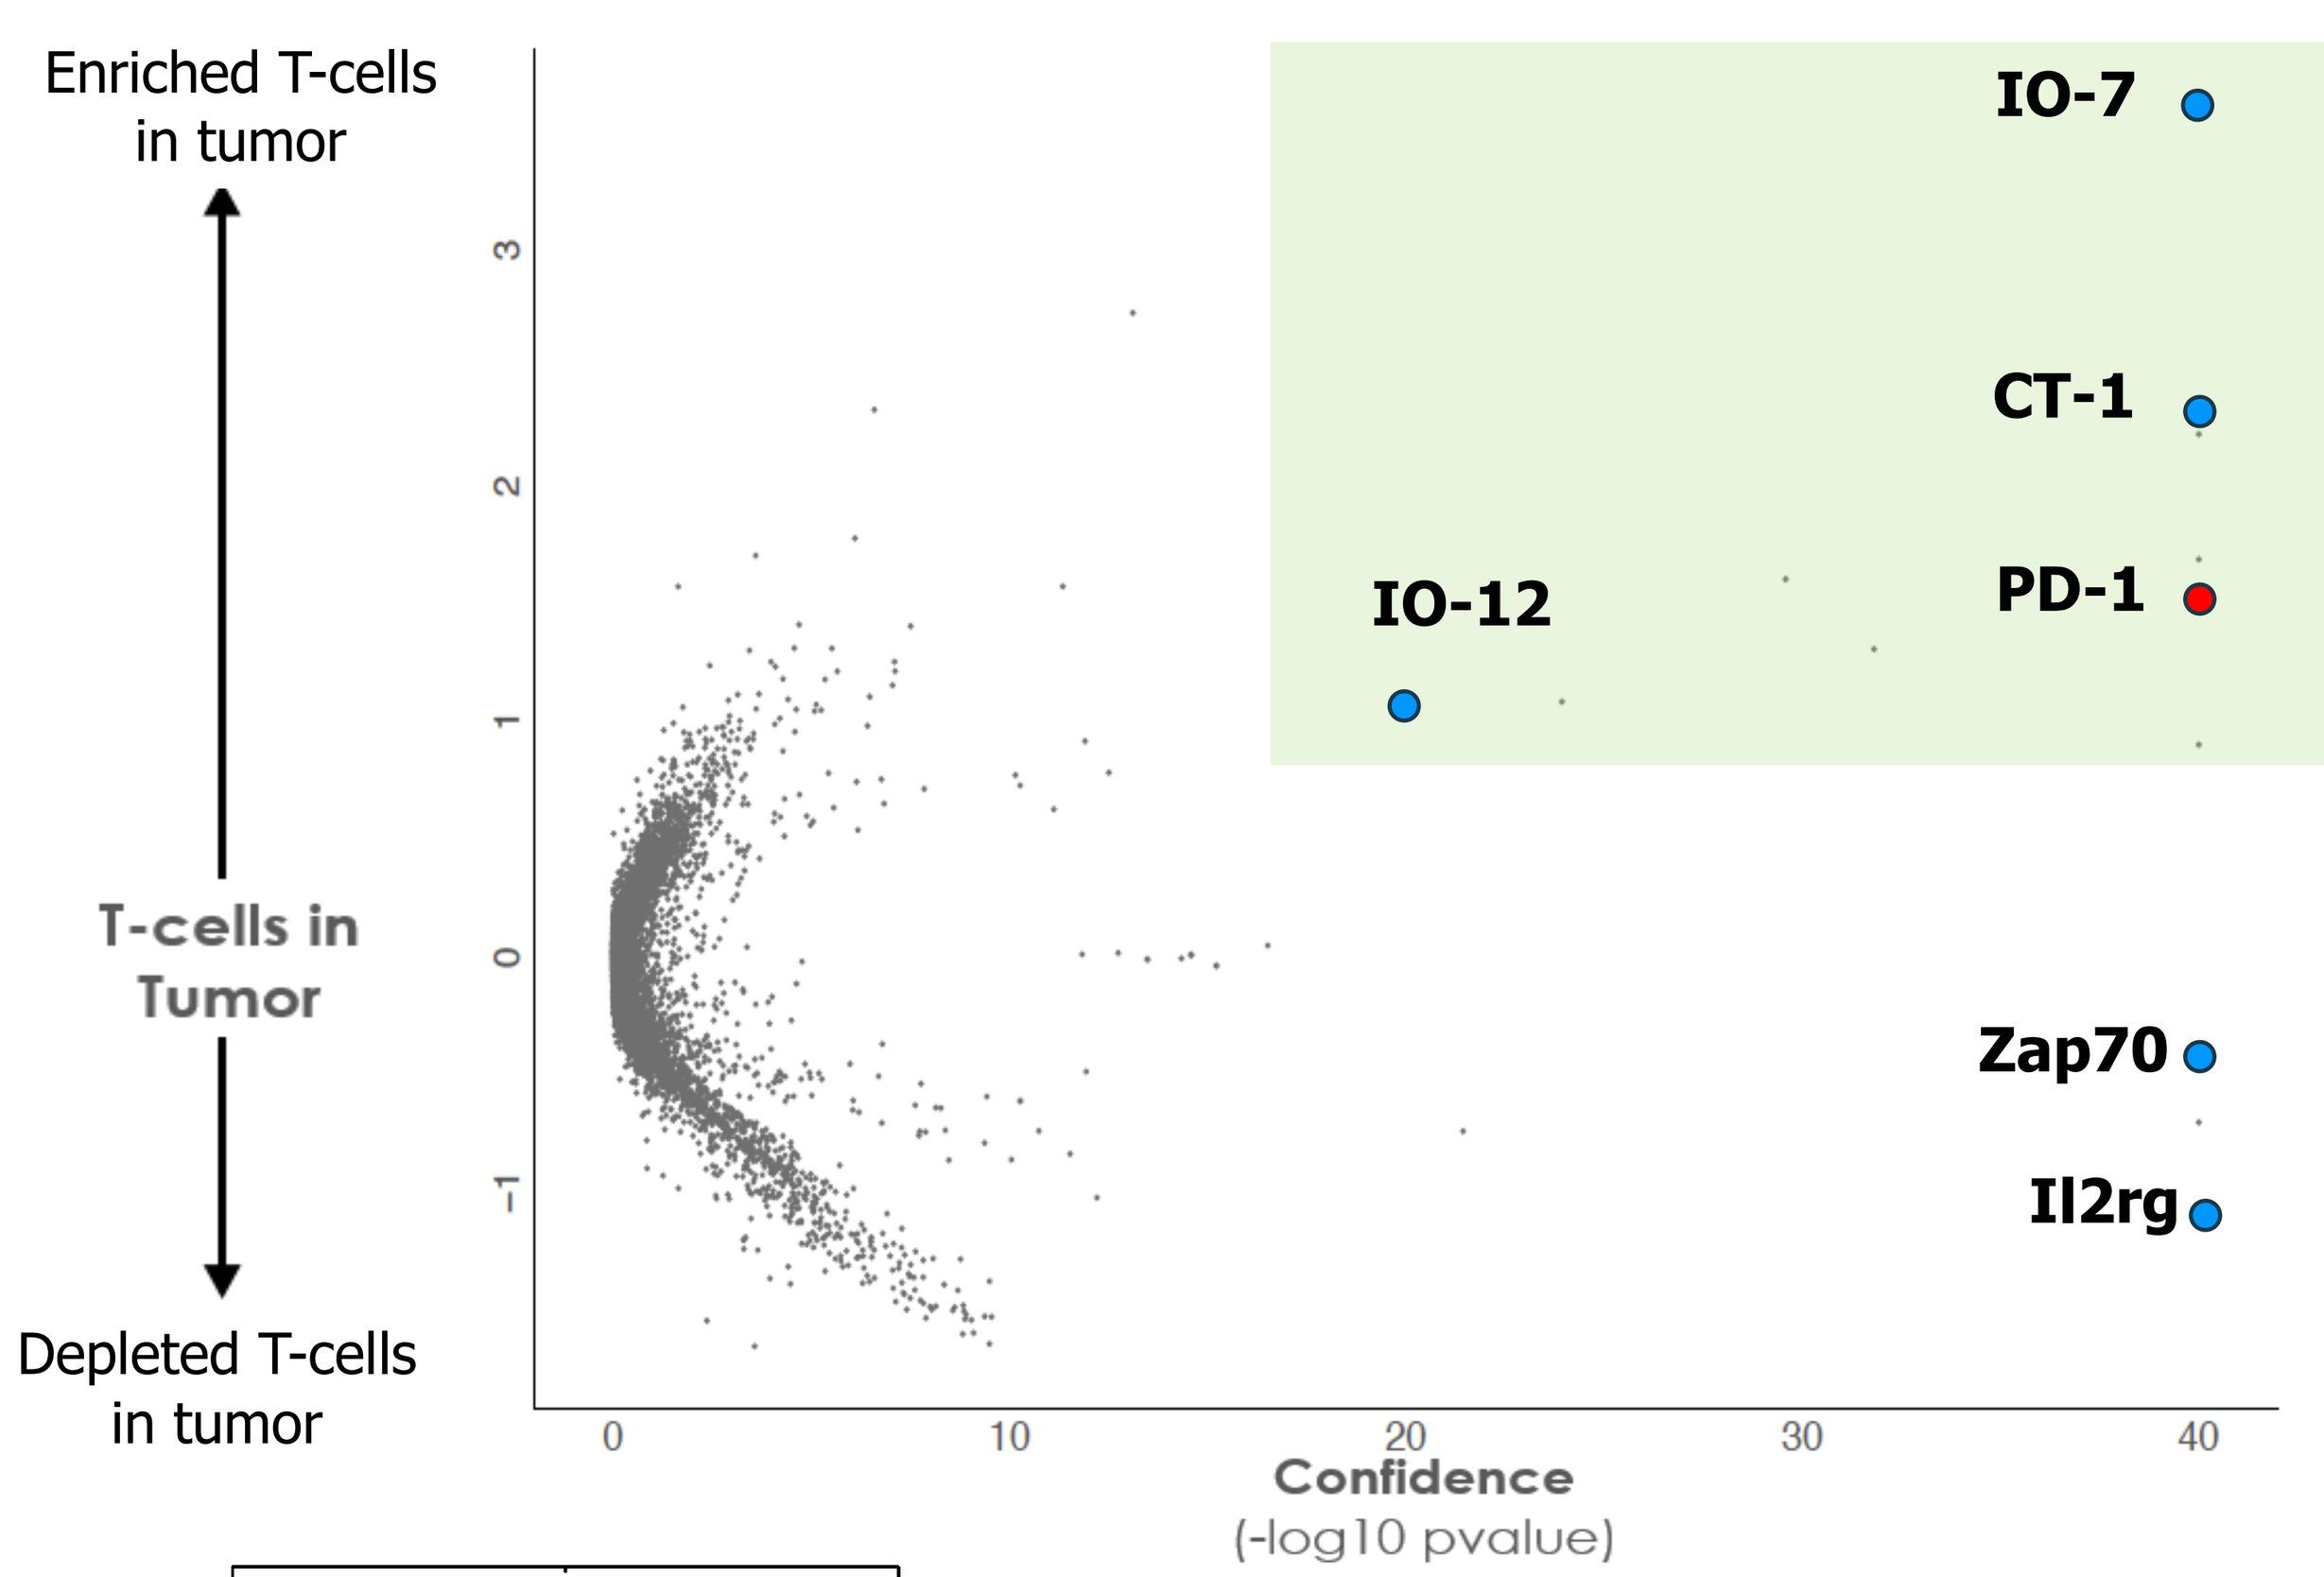
Enriched T-cells in tumor
T-cells in Tumor
Depleted T-cells in tumor
IO-7
CT-1
IO-12
PD-1
Zap70
Il2rg
0
10
20
30
40
Confidence
(-log10 pvalue)

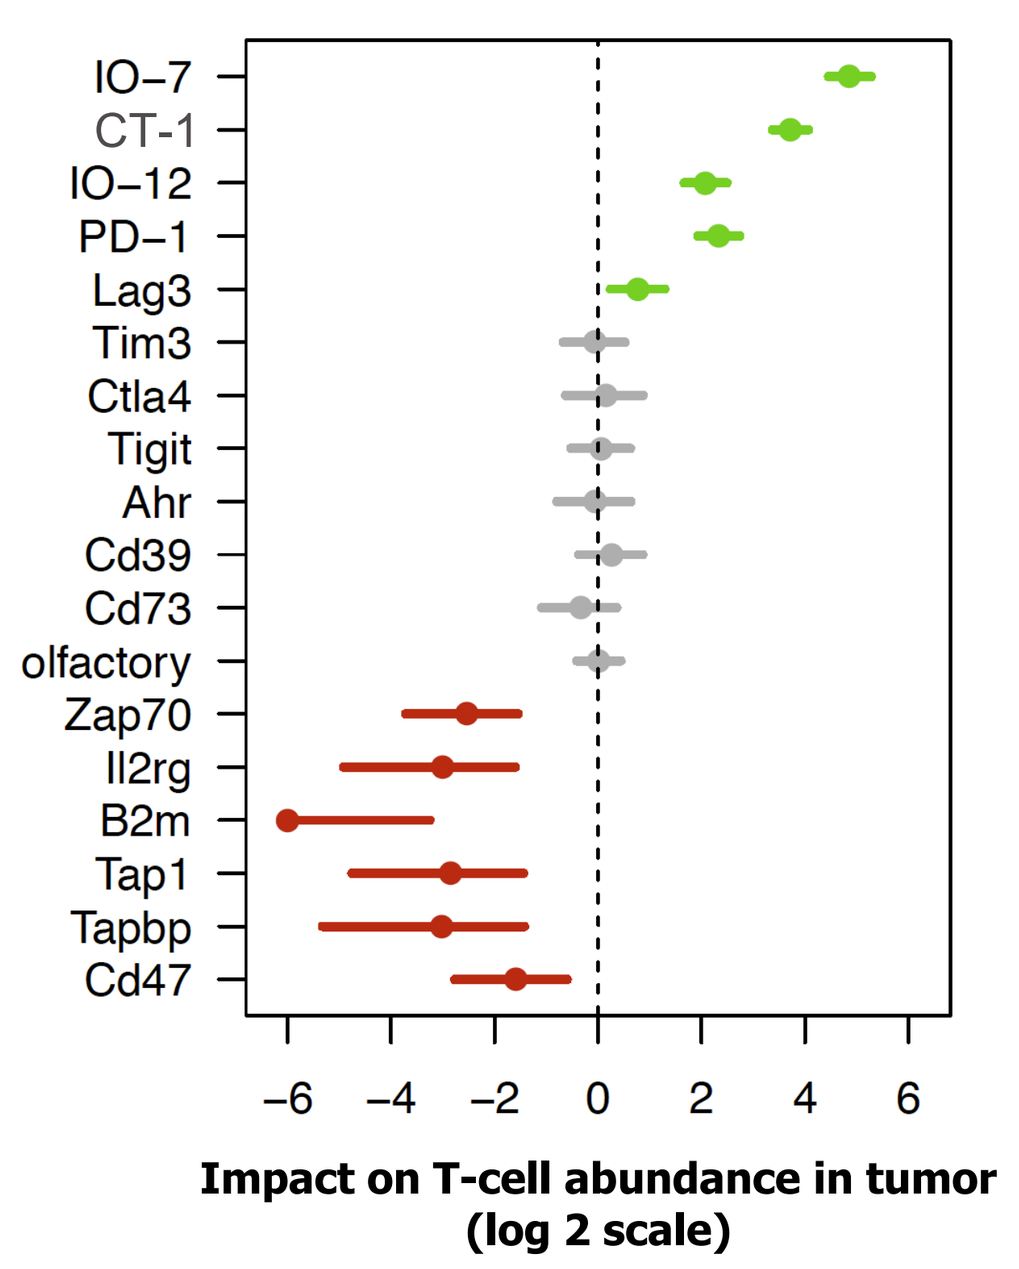
IO–7
CT–1
IO–12
PD–1
Lag3
Tim3
Ctla4
Tigit
Ahr
Cd39
Cd73
olfactory
Zap70
Il2rg
B2m
Tap1
Tapbp
Cd47
−6
−4
−2
0
2
4
6
Impact on T-cell abundance in tumor (log 2 scale)

- Multiple targets identified that drive T cell enrichment similar or greater than PD-1 (green box above)
- PD-1 serves as a positive control. IO-7, CT-1 and IO-12 are KSQ portfolio targets.
- Critical components of T-cell activation (Zap70) and survival pathways (Il2rg) are identified as depleted targets in the screen (left graph)

Figure 2: Novel targets drive T cell enrichment across PD-1 sensitive and insensitive models

Novel T cell targets also score in a PD-1 insensitive PMEL / gp100 CRC model

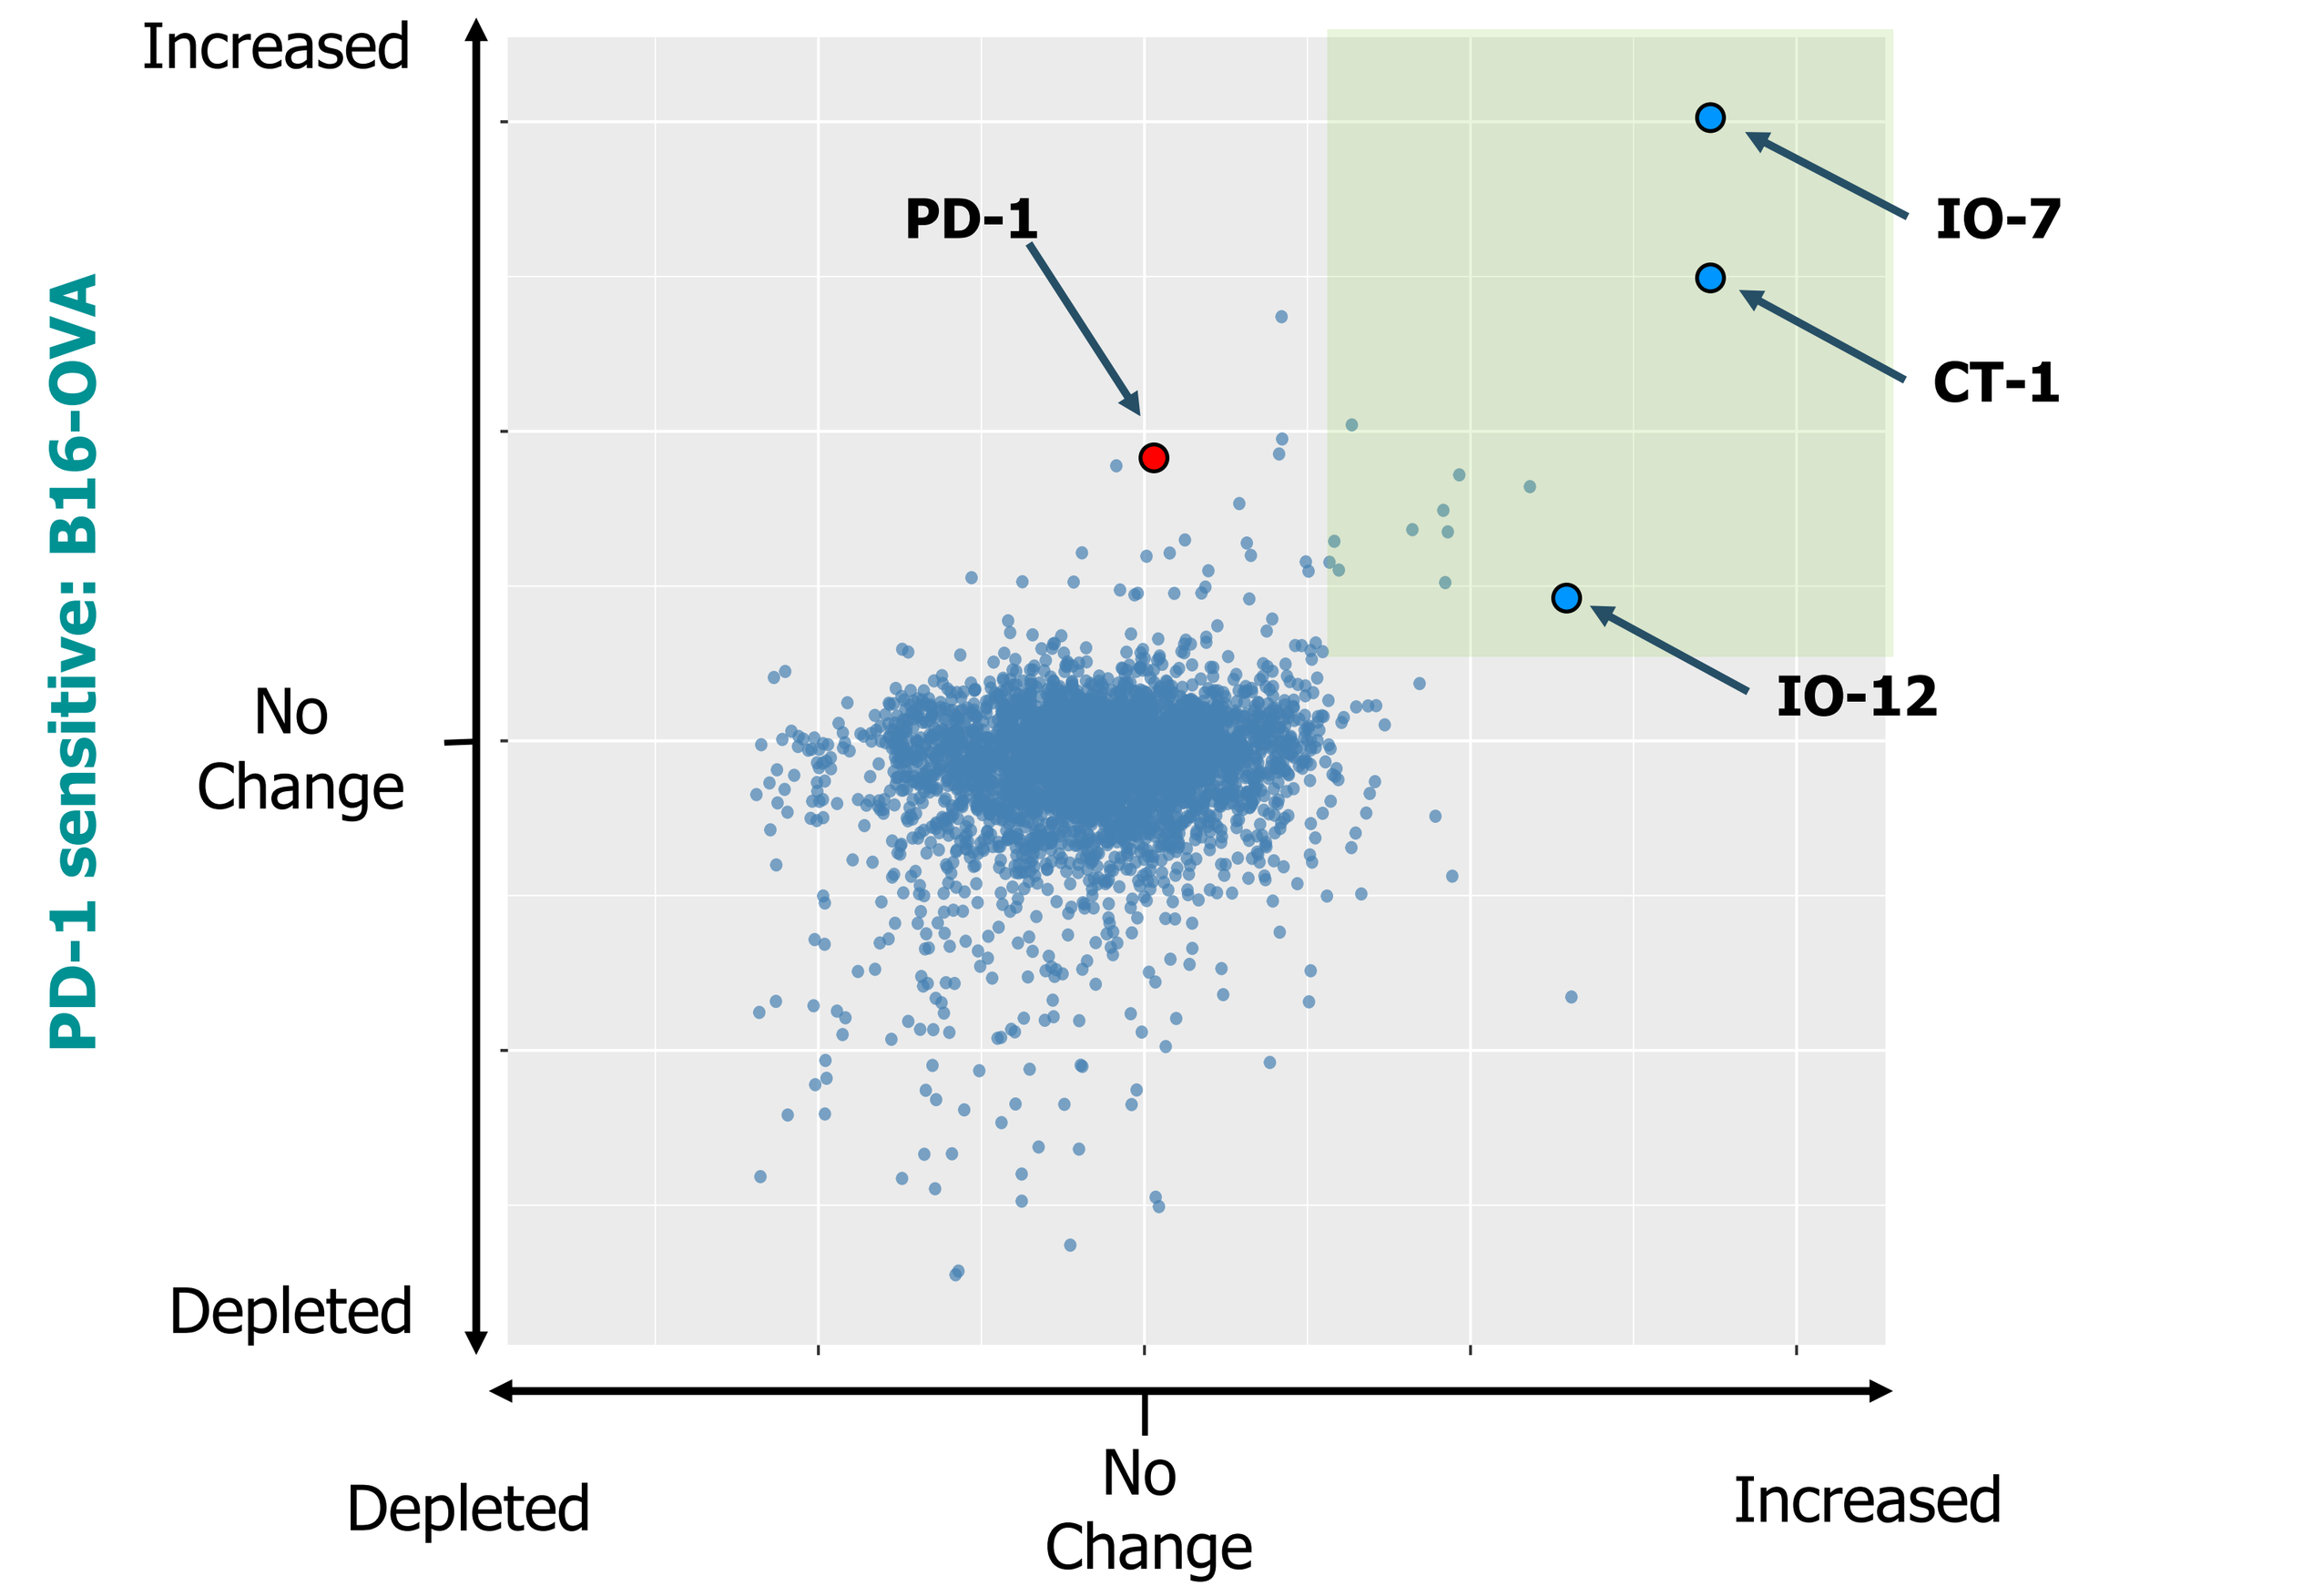
PD-1 sensitive: B16-OVA
Increased
No Change
Depleted
PD-1
IO-7
CT-1
IO-12
Depleted
No Change
Increased
PD-1 insensitive: gp100 CRC

- IO-7, CT-1 and IO-12 sgRNAs were also found to drive T cell enrichment in a PMEL / gp100 CRC tumor CRISPR screen (green box)
- PD-1 does not score in the PMEL / gp100 CRC screen, consistent with this model being PD-1 insensitive

Figure 3: KSQ Targets IO-7, CT-1 and IO-12 demonstrate robust efficacy in PD-1 sensitive syngeneic tumor models

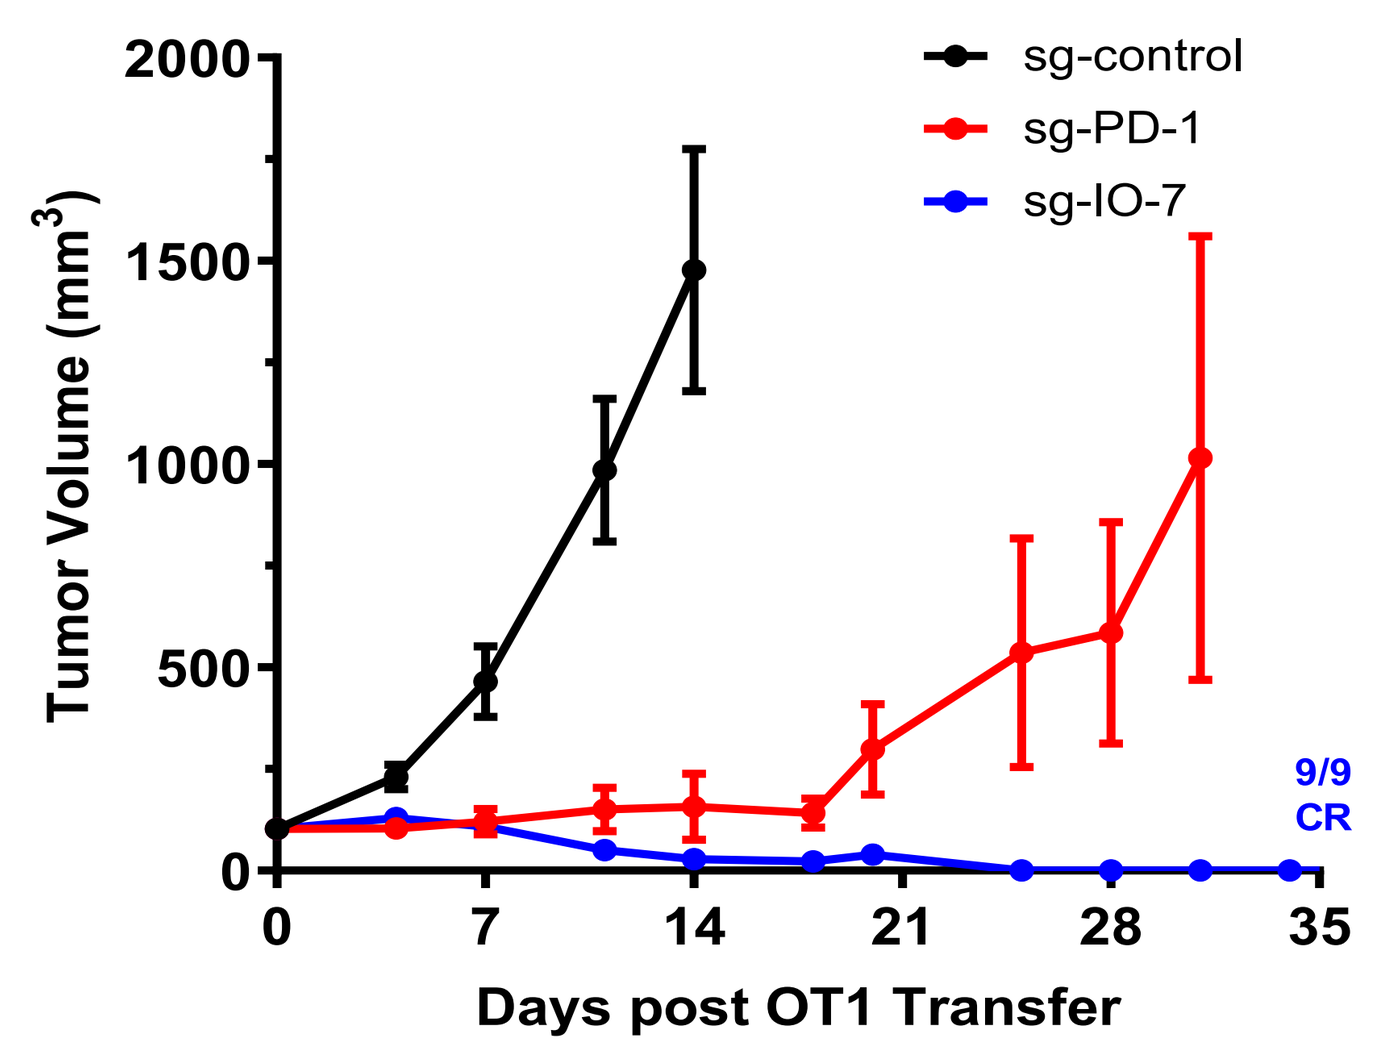
sg-control
sg-PD-1
sg-IO-7
Tumor Volume (mm³)
9/9 CR
Days post OT1 Transfer

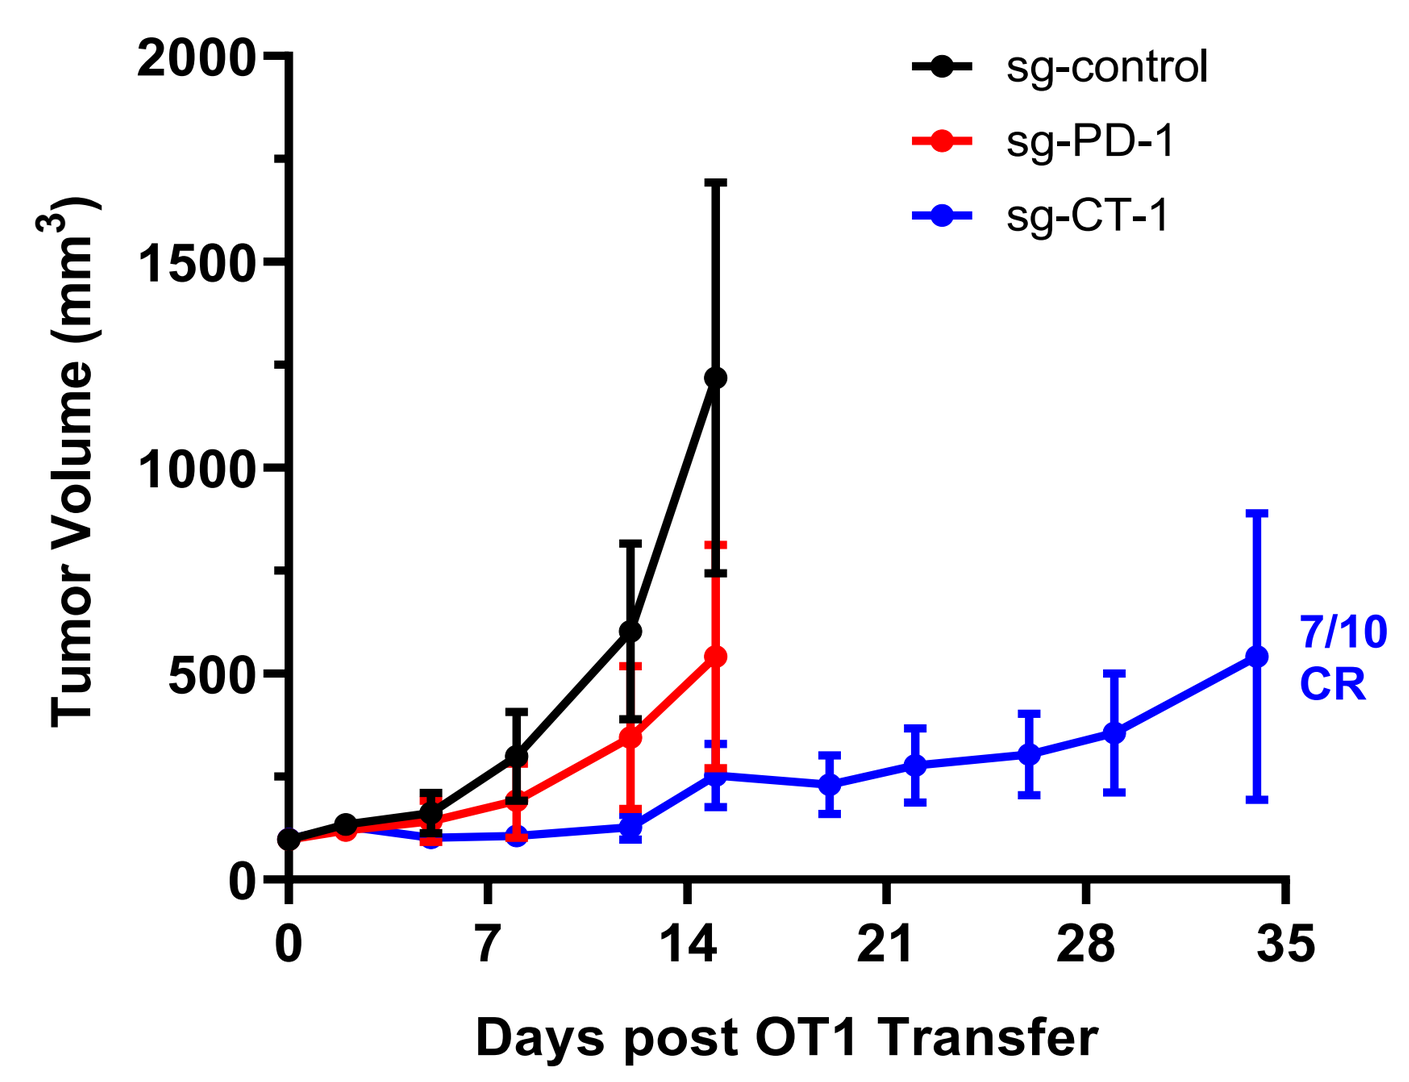
sg-control
sg-PD-1
sg-CT-1
Tumor Volume (mm³)
7/10 CR
Days post OT1 Transfer

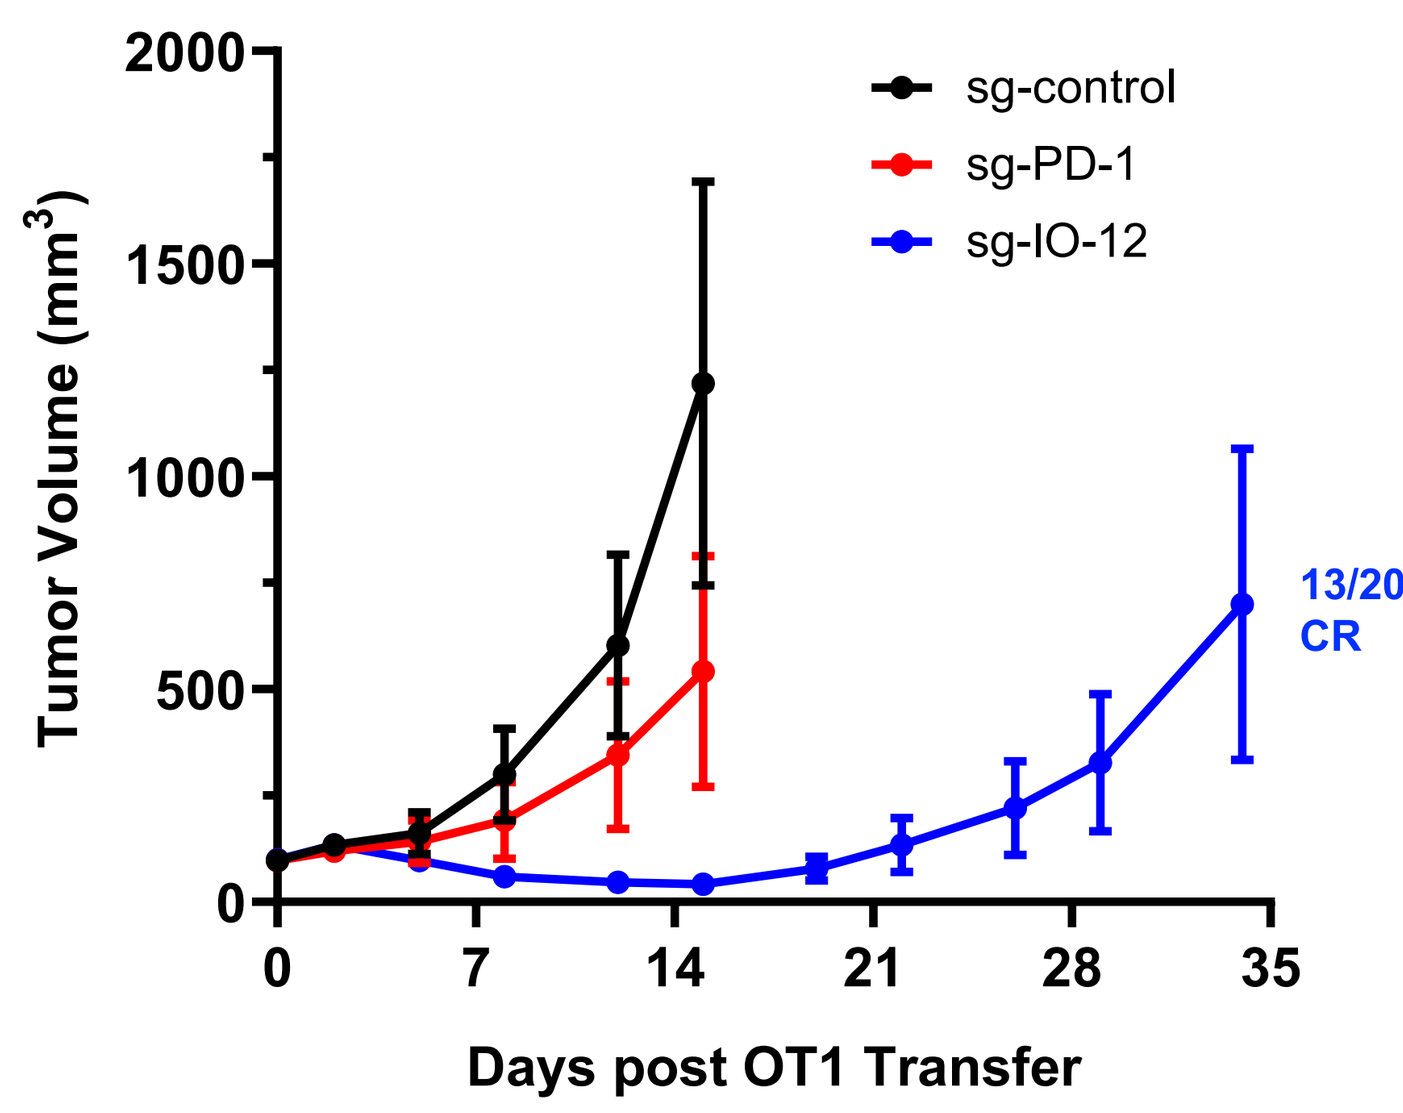
sg-control
sg-PD-1
sg-IO-12
Tumor Volume (mm³)
13/20 CR
Days post OT1 Transfer

- Adoptive transfer of IO-7, CT-1 and IO-12-edited OT1 T cells into mice bearing established 100mm³ B16-OVA tumors demonstrate efficacy greater than PD-1-edited OT1 T cells

Figure 4: KSQ Targets IO-7 and CT-1 demonstrate robust efficacy in PD-1 insensitive syngeneic tumor models

B16-F10 Lung Metastatic model

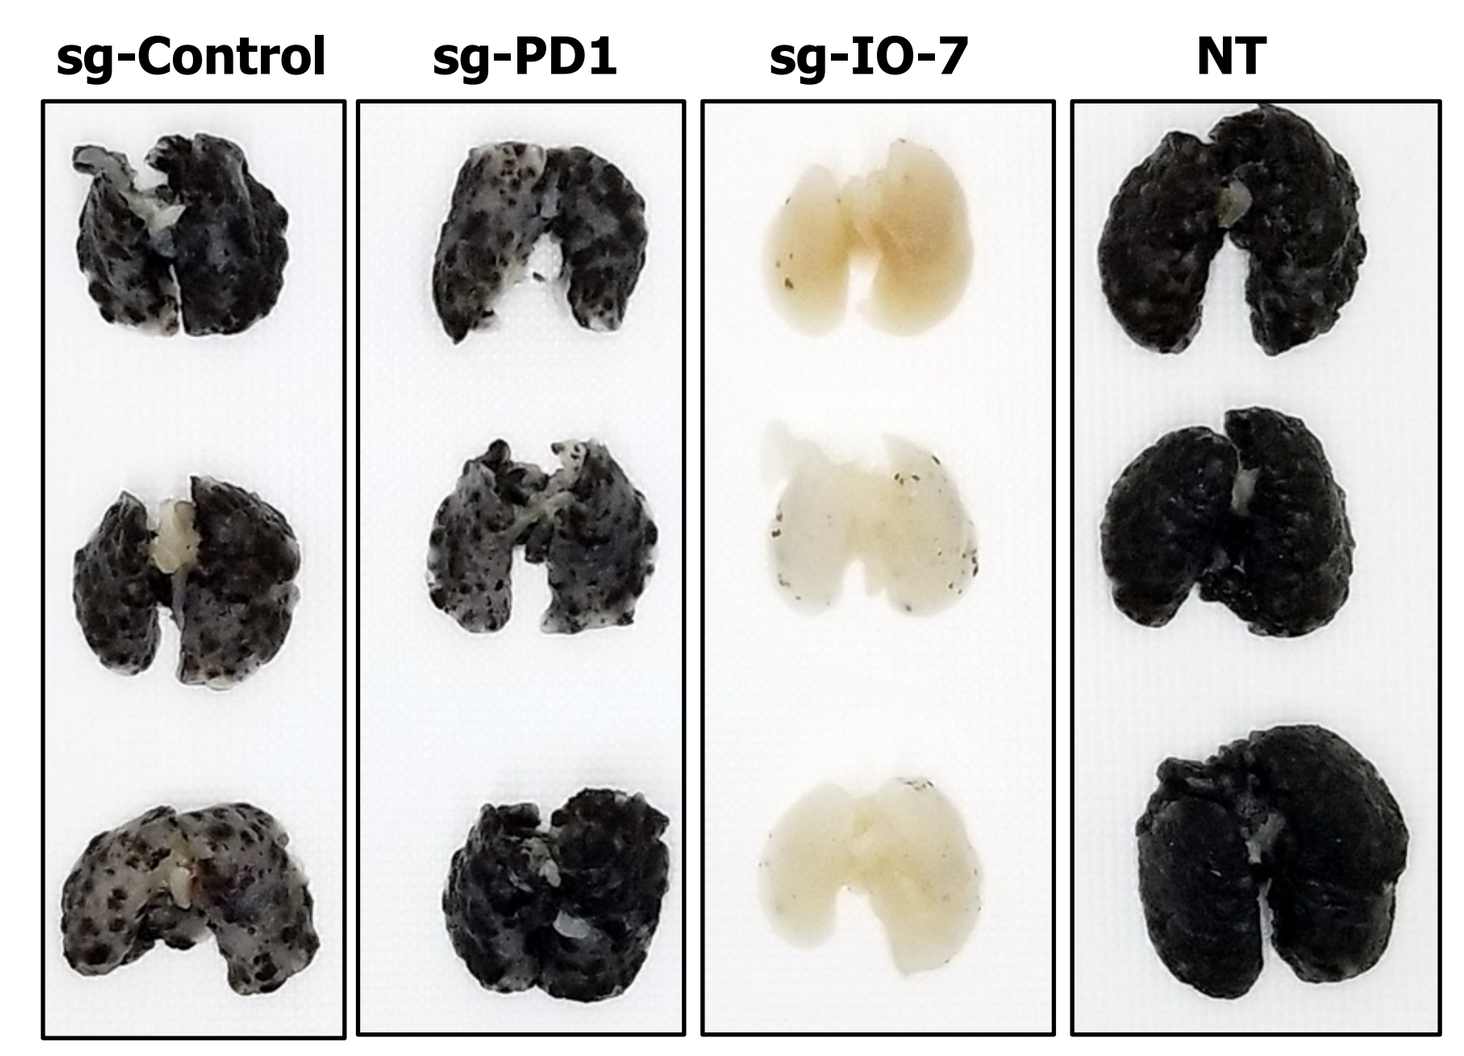
sg-Control
sg-PD1
sg-IO-7
NT

gp100 CRC

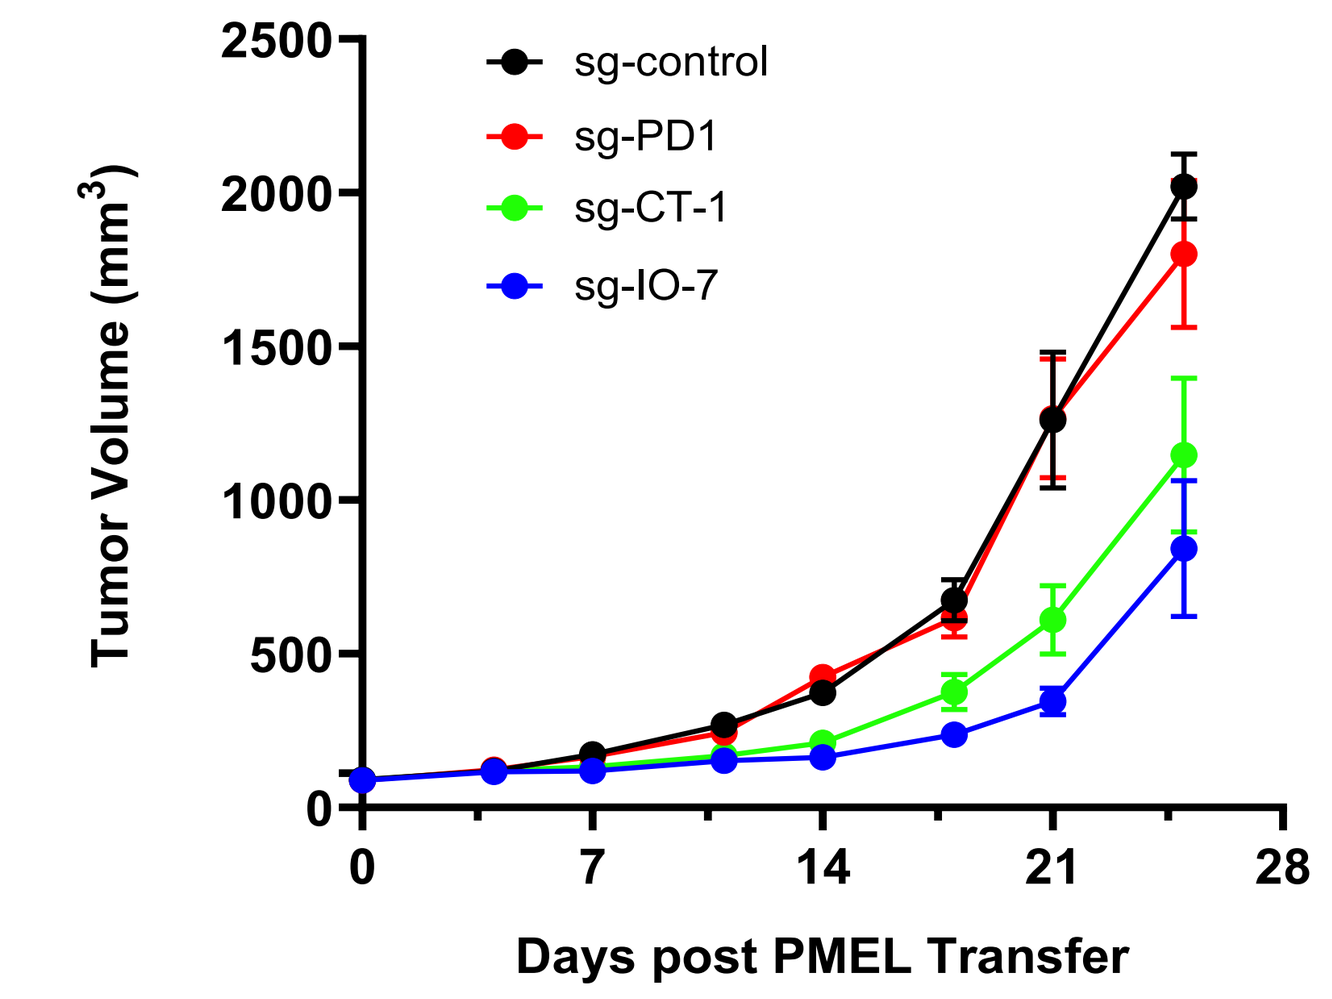
sg-control
sg-PD1
sg-CT-1
sg-IO-7
Tumor Volume (mm³)
Days post PMEL Transfer

B16-OVA Large Tumors

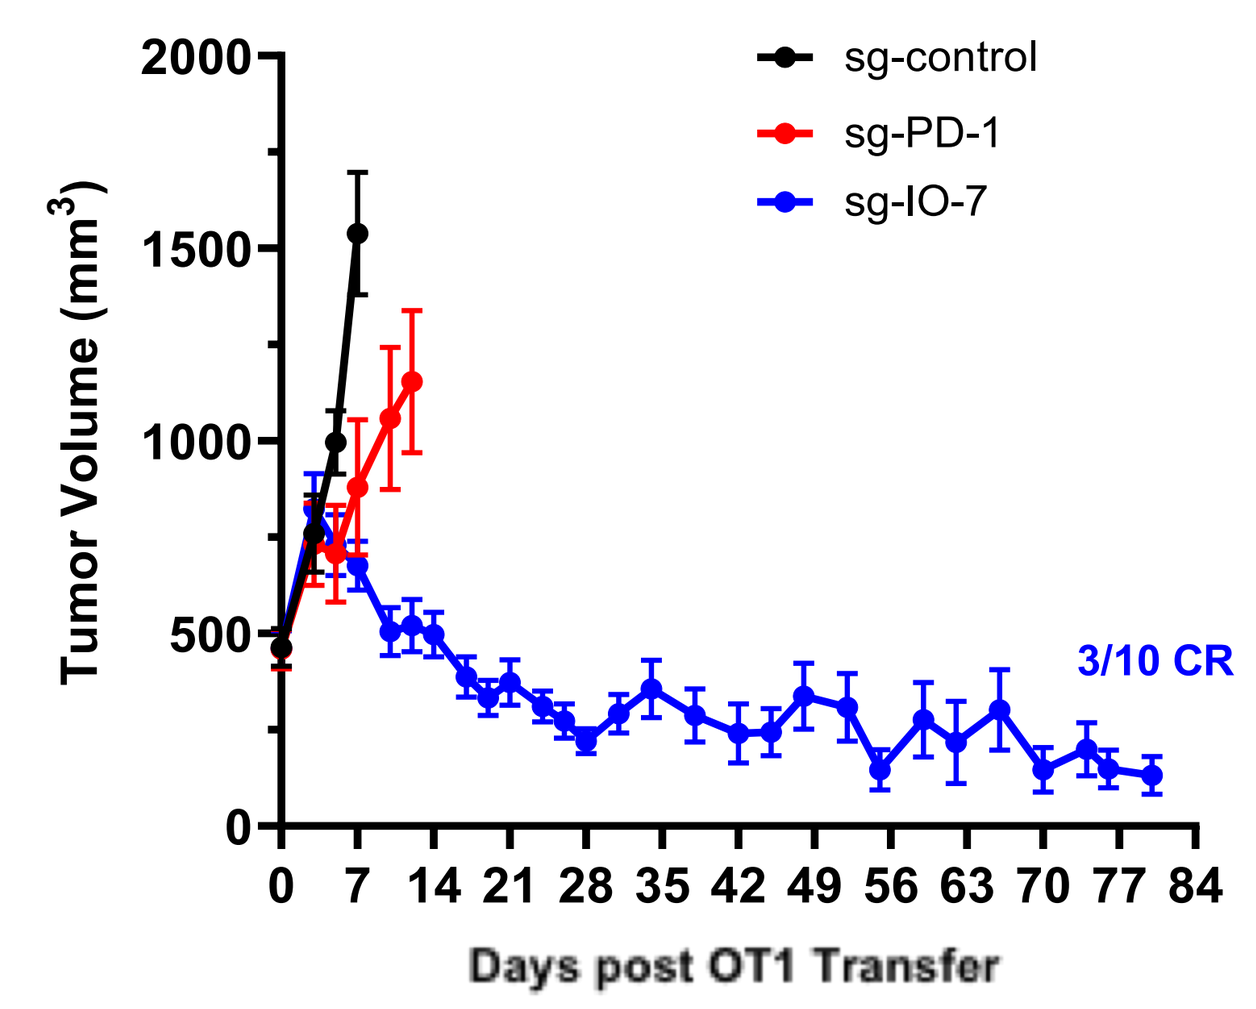
sg-control
sg-PD-1
sg-IO-7
Tumor Volume (mm³)
3/10 CR
Days post OT1 Transfer

- Transfer of IO-7 edited PMEL T cells show therapeutic efficacy in B16-F10 lung met model
- IO-7 and CT-1 edited PMEL T cells show efficacy in a gp100 CRC model
- Transfer of IO-7 edited OT1 T cells into mice with established 450mm³ tumors demonstrate robust efficacy
- PD-1 edited T cells have no activity in these models

Figure 5: scRNA-SEQ profiling of tumor shows IO-7 edited T cells drive expansion of CD8 Tscm cells

IO-7 edited OT1 drive expansion of CD8 Tcm cells and depletion of Tregs

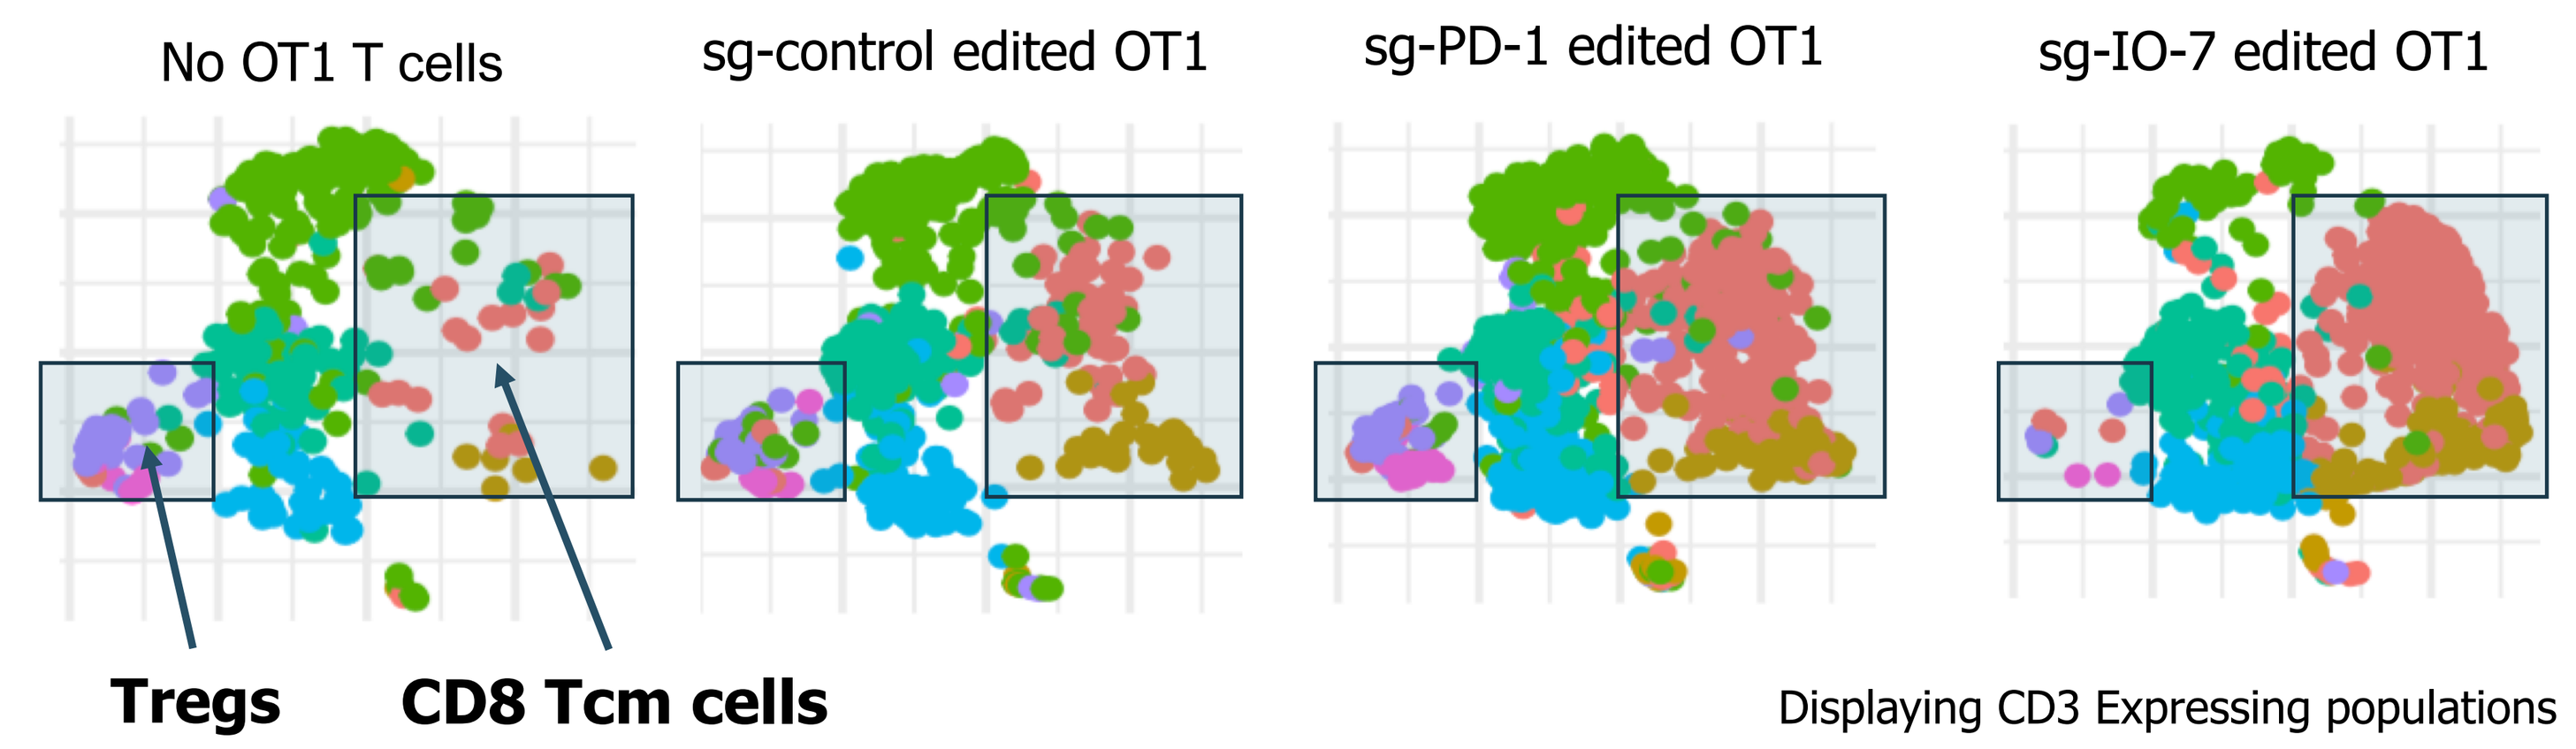
No OT1 T cells
sg-control edited OT1
sg-PD-1 edited OT1
sg-IO-7 edited OT1
Tregs
CD8 Tcm cells

Displaying CD3 Expressing populations

CD8 Tcm cells express TCF7 and resemble Tscm cells

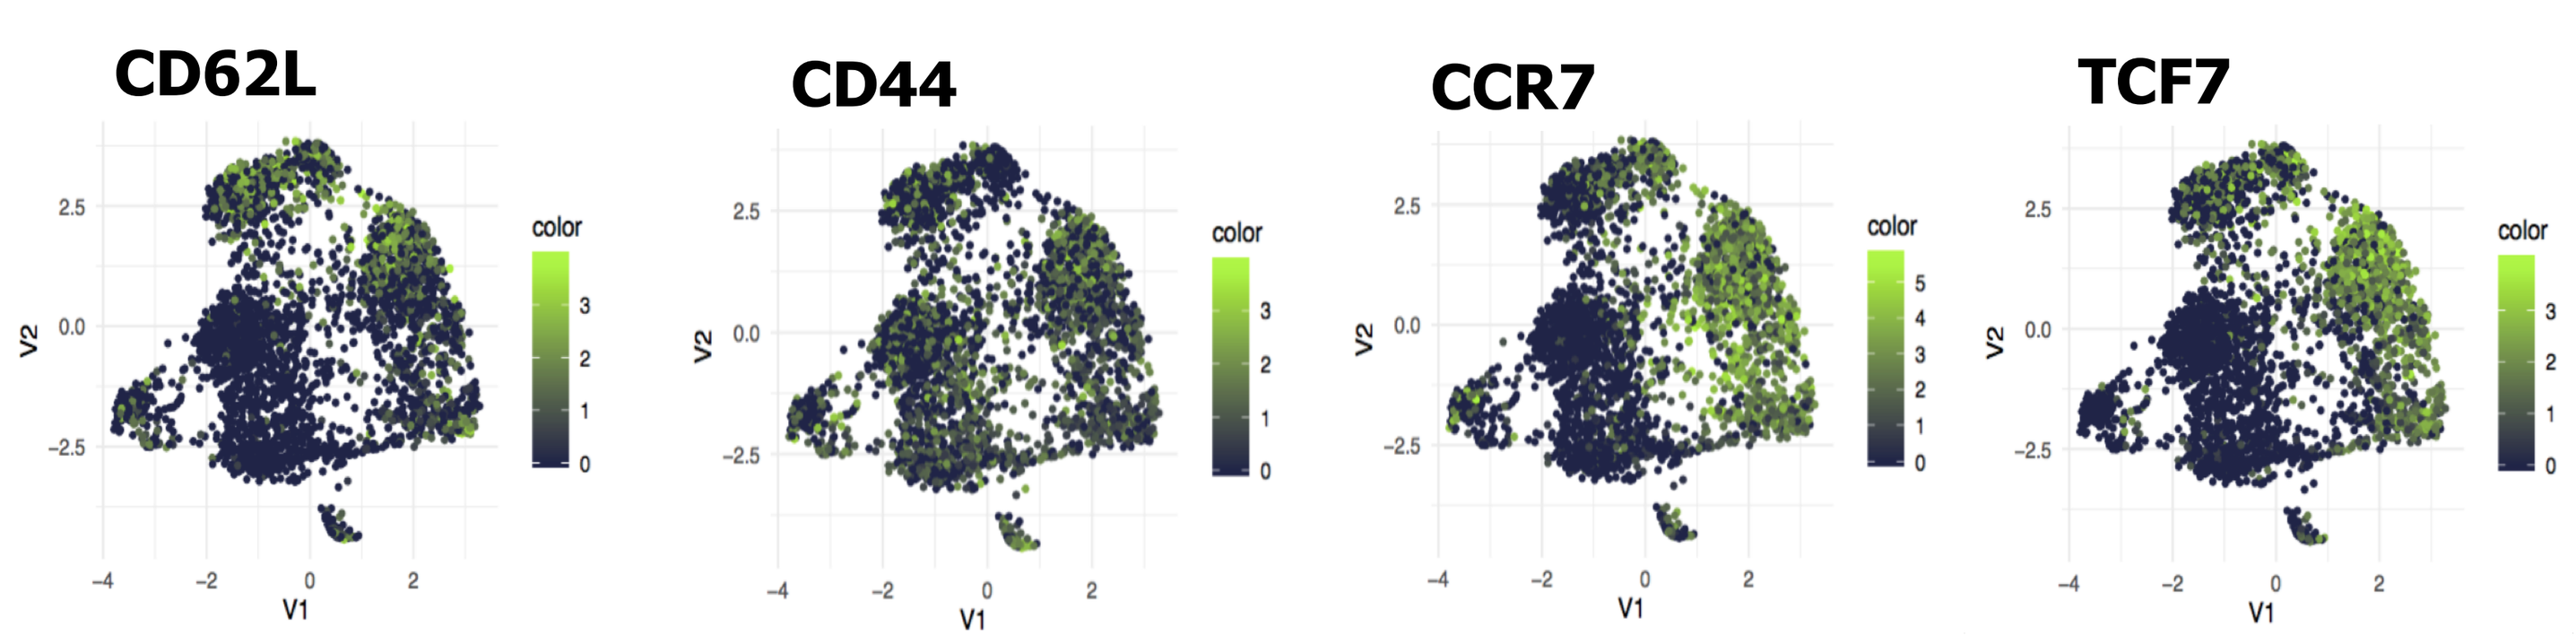
CD62L
CD44
CCR7
TCF7
V1
V2
color

- Adoptive transfer of IO-7 edited OT1 into tumor bearing mice expand a CCR7⁺TCF7⁺ Tscm population and deplete Tregs from the tumor microenvironment

Conclusions

- An in vivo genome-wide CRISPR T-cell screen identifies multiple targets scoring similarly or greater than PD-1 in multiple syngeneic models
- Inhibition of IO-7, CT-1 and IO-12 in CD8 T cells translated to robust activity across multiple syngeneic models where PD-1 inhibition showed no activity
- **KSQ is developing novel therapeutics against in vivo validated targets for the treatment of checkpoint-inhibitor-refractory patients**